Poster 599

Patient-Reported Injection-Site Pain and Treatment Satisfaction After Switching From Long-Term Prophylaxis to Donidalorsen for the Treatment of Hereditary Angioedema: Results From the OASISplus Study

Marc A Riedl[1], Jonathan A Bernstein[2], Raffi Tachdjian[3], Aaron Yarlas[4], Laura Bordone[4], Kenneth B Newman[4], Sabrina Treadwell[4], Tao Lin[4], Danny M Cohn[5]

[1]Division of Allergy and Immunology, University of California San Diego School of Medicine, La Jolla, CA, USA; [2]Division of Rheumatology, Allergy, and Immunology, Department of Internal Medicine, University of Cincinnati College of Medicine, Cincinnati, OH, USA; [3]Division of Allergy, Immunology, and Rheumatology, University of California Los Angeles, Los Angeles, CA, USA; [4]Ionis, Carlsbad, CA, USA; [5]Department of Vascular Medicine, Amsterdam Cardiovascular Sciences, Amsterdam UMC, University of Amsterdam, Amsterdam, the Netherlands

Background

- Hereditary angioedema (HAE) is a rare disease characterized by frequently severe, potentially life-threatening attacks of tissue swelling[1,2]
- HAE is most often caused by C1 inhibitor (C1INH) deficiency (HAE-C1INH-Type1) or dysfunction (HAE-C1INH-Type2)[3]
- Donidalorsen is an investigational RNA-targeted antisense oligonucleotide that specifically reduces prekallikrein production in the liver[4]
- In the pivotal phase 3 OASIS-HAE study, donidalorsen 80 mg subcutaneously (SC) once every 4 weeks (Q4W) or every 8 weeks (Q8W) significantly reduced HAE attack rate and improved patient-reported quality of life[4]

Study Design

Figure 1. Study Design

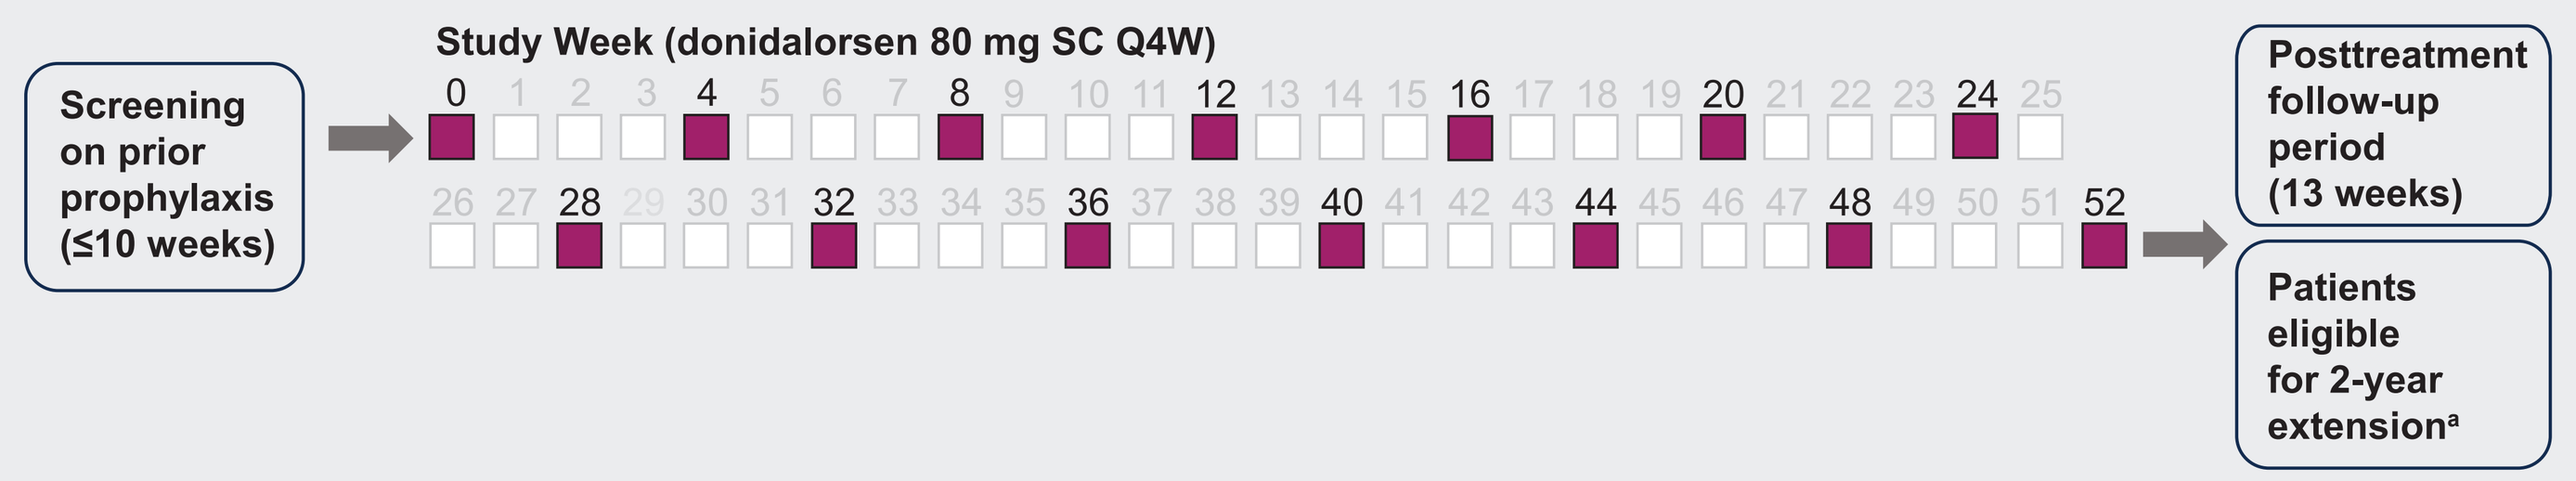

Study Week (donidalorsen 80 mg SC Q4W)
Screening on prior prophylaxis (≤10 weeks)
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25
26 27 28 29 30 31 32 33 34 35 36 37 38 39 40 41 42 43 44 45 46 47 48 49 50 51 52
Posttreatment follow-up period (13 weeks)
Patients eligible for 2-year extension[a]

[a]Patients could switch to Q8W dosing after Year 1.
Q4W, once every 4 weeks; Q8W, once every 8 weeks; SC, subcutaneously.

- In the ongoing phase 3 OASISplus (NCT05392114) study, the Switch cohort included patients ≥12 years of age with HAE-C1INH-Type1 or HAE-C1INH-Type2 on stable doses of lanadelumab, berotralstat, or C1INH for ≥12 weeks
- All patients switched to donidalorsen 80 mg SC Q4W with no washout period
- Patients received the last dose of their prior long-term prophylactic medication (LTP) based on a prespecified algorithm as follows:
 - Lanadelumab: 14 ± 3 days before day 1
 - Berotralstat: 14 ± 3 days after day 1
 - C1INH: 14 ± 3 days after day 1
- Endpoints included:
 - Safety over Weeks 0 to 16
 - Incidence and severity of treatment-emergent adverse events (primary endpoint)
 - Injection-site pain (ISP) at baseline and Week 16
 - ISP was measured on an 11-point numeric rating scale (values from 0–10) with 0 indicating "no pain," 5 indicating "moderate pain," and 10 indicating "worst possible pain"
 - Treatment satisfaction assessed by Treatment Satisfaction Questionnaire for Medication, version 2 (TSQM-II)[5] at baseline and Week 16
 - TSQM-II yields scores in 4 domains: effectiveness, side effects, convenience, global satisfaction
 - All scores are on a 0 to 100 scale with higher scores indicating greater satisfaction
 - Treatment preference at Week 16
 - Treatment Preference Assessment was a self-reported measure of preference for current or previous treatment
 - Treatment preference was measured on a 5-point preference scale with "strong preference" for current or previous treatment on both extremes of the scale, and "no preference" in the middle
 - If a patient preferred one treatment over the other, they were asked for a reason from a list and were given the option to choose "other" to give an open-ended response
- Data are shown from an interim February 2024 data cutoff

Results

Table 1. Patient Disposition and Demographics

	Patients switching to donidalorsen 80 mg Q4W from:			
	Lanadelumab	Berotralstat	C1INH	Total
Patients enrolled, n	32	11	22	65
Patients dosed, n	31	11	22	64
Completed Week 16 of treatment, n (%)	28 (88)	10 (91)	20 (91)	58 (89)
Early termination, n (%)				
Lack of efficacy	3 (9)	0	1 (5)	4 (6)
Adverse event	1 (3)	0	0	1 (2)
Lost to follow-up	1 (3)	0	0	1 (2)
Voluntary withdrawal	0	0	1 (5)	1 (2)
Other (not dosed)	1 (3)	0	0	1 (2)
Age, years, mean (SD)	40 (14)	46 (11)	41 (17)	42 (15)
Age, years, n (%)				
12–17 years of age	1 (3)	0	3 (14)	4 (6)
≥18 years of age	30 (97)	11 (100)	19 (86)	60 (94)
Sex, n (%)				
Male	17 (55)	3 (27)	6 (27)	26 (41)
Female	14 (45)	8 (73)	16 (73)	38 (59)
Race,[a] n (%)				
White	26 (84)	11 (100)	20 (91)	57 (89)
Multiple or other[b]	5 (16)	0	2 (9)	7 (11)

[a]Race was self-reported by patients during screening. [b]Includes American Indian or Alaska Native, Asian, Black or African American, Native Hawaiian or Other Pacific Islander, multiple races, and other races.
C1INH, C1 inhibitor; Q4W, once every 4 weeks; SD, standard deviation.

- As of February 28, 2024, 58 of 64 dosed patients (89%) had completed Week 16 of treatment
- Mean exposure to donidalorsen was 263 days

Table 2. Safety at Week 16

	Patients switching to donidalorsen 80 mg Q4W from:		
	Lanadelumab (n = 31)	Berotralstat (n = 11)	C1INH (n = 22)
Any TEAE,[a] n (%)	22 (71.0)	8 (72.7)	15 (68.2)
Related to study drug	9 (29.0)	4 (36.4)	4 (18.2)
Leading to discontinuation	1[b] (3.2)	0	0
Severity of TEAEs, n (%)			
Mild	5 (16.1)	6 (54.5)	6 (27.3)
Moderate	15 (48.4)	1 (9.1)	9 (40.9)
Severe	2 (6.5)	1 (9.1)	0
Any SAE	1[b] (3.2)	0	0
Injection-site reactions (≥5% in any group), n (%)			
Erythema	3 (9.7)	1 (9.1)	3 (13.6)
Pruritus	3 (9.7)	2 (18.2)	0
Pain	3 (9.7)	0	1 (4.5)
Bruising	1 (3.2)	1 (9.1)	0

[a]TEAE is defined as any adverse event starting or worsening on or after the first dose of donidalorsen. [b]One patient discontinued due to a series of cardiac failure and renal disorder TEAEs, including serious TEAEs of heart failure exacerbation and renal infarct, that were not related to the study drug.
C1INH, C1 inhibitor; Q4W, once every 4 weeks; SAE, serious adverse event; TEAE, treatment-emergent adverse event.

- All injection-site reactions were mild or moderate in severity

Figure 2. Injection-Site Pain Score at Baseline and Week 16

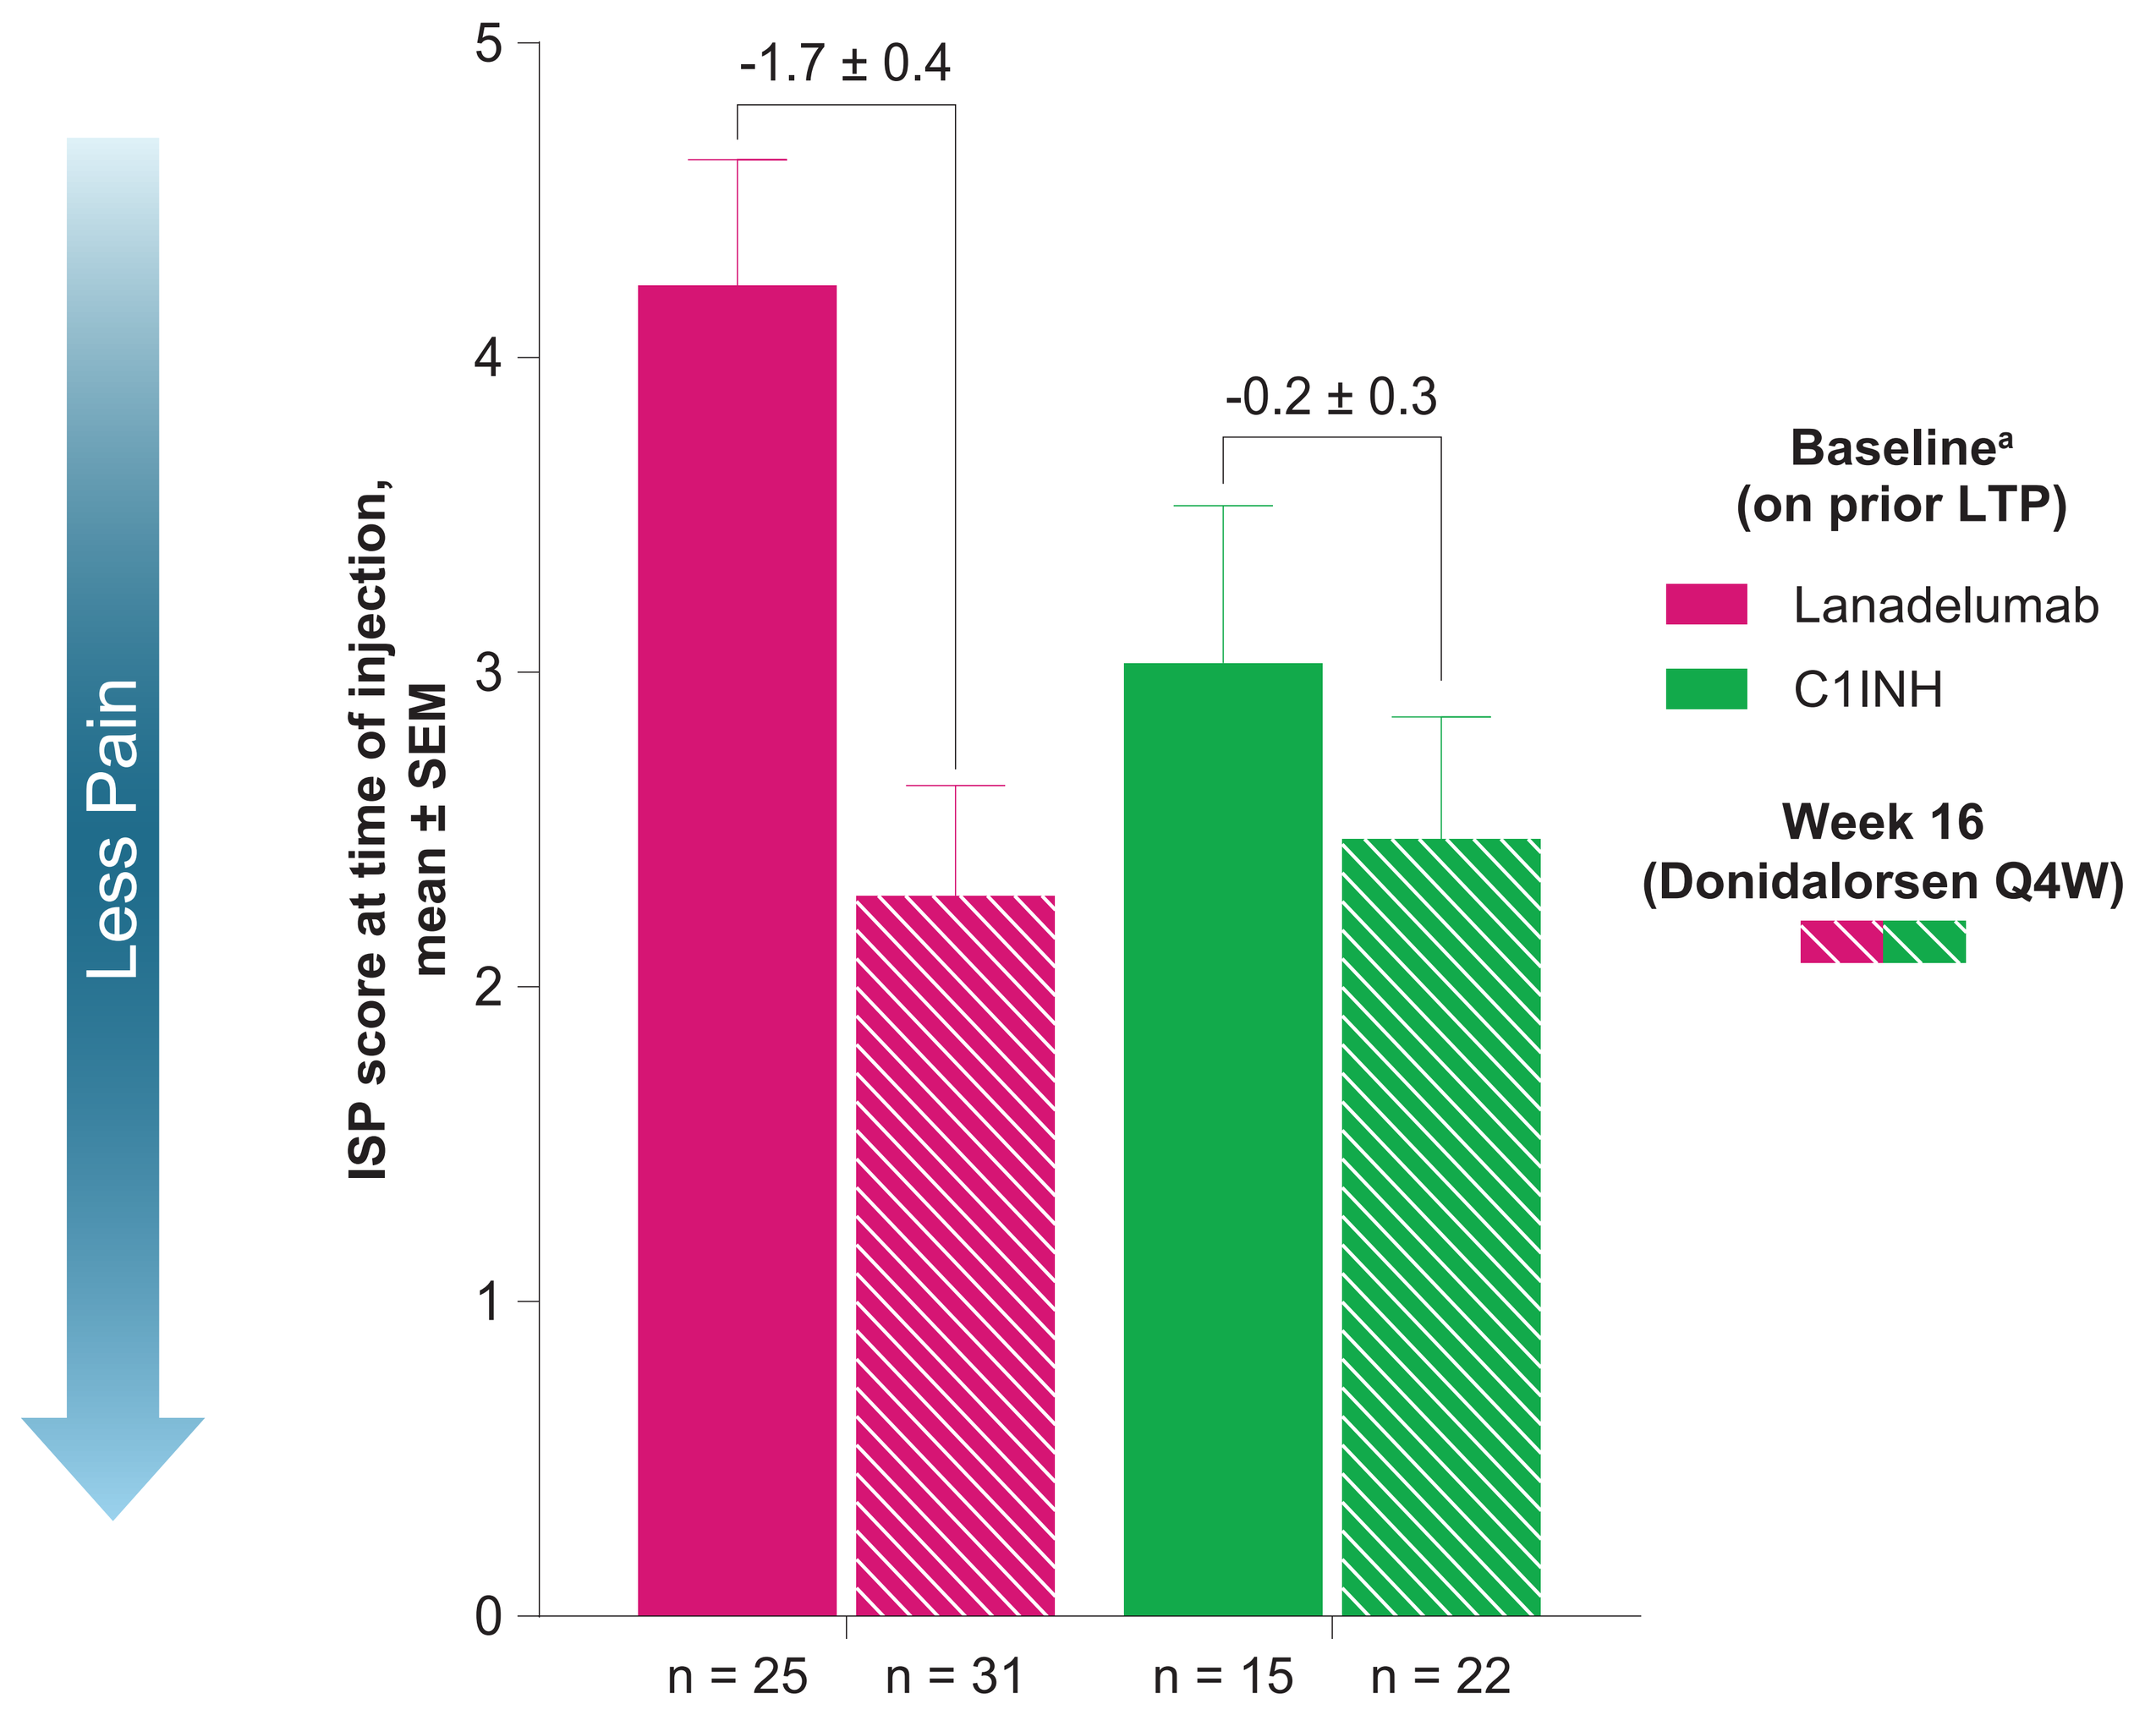

Less Pain
ISP score at time of injection, mean ± SEM
0
1
2
3
4
5
-1.7 ± 0.4
-0.2 ± 0.3
n = 25
n = 31
n = 15
n = 22
Baseline[a] (on prior LTP)
Lanadelumab
C1INH
Week 16 (Donidalorsen Q4W)

[a]Week 0 in the OASISplus study.
C1INH, C1 inhibitor; ISP, injection-site pain; LTP, long-term prophylactic medication; Q4W, once every 4 weeks; SEM, standard error of the mean.

- Overall, mean ISP scores decreased by 1.2 points after switching from prior injectable LTP to donidalorsen

Figure 3. Treatment Preference at Week 16

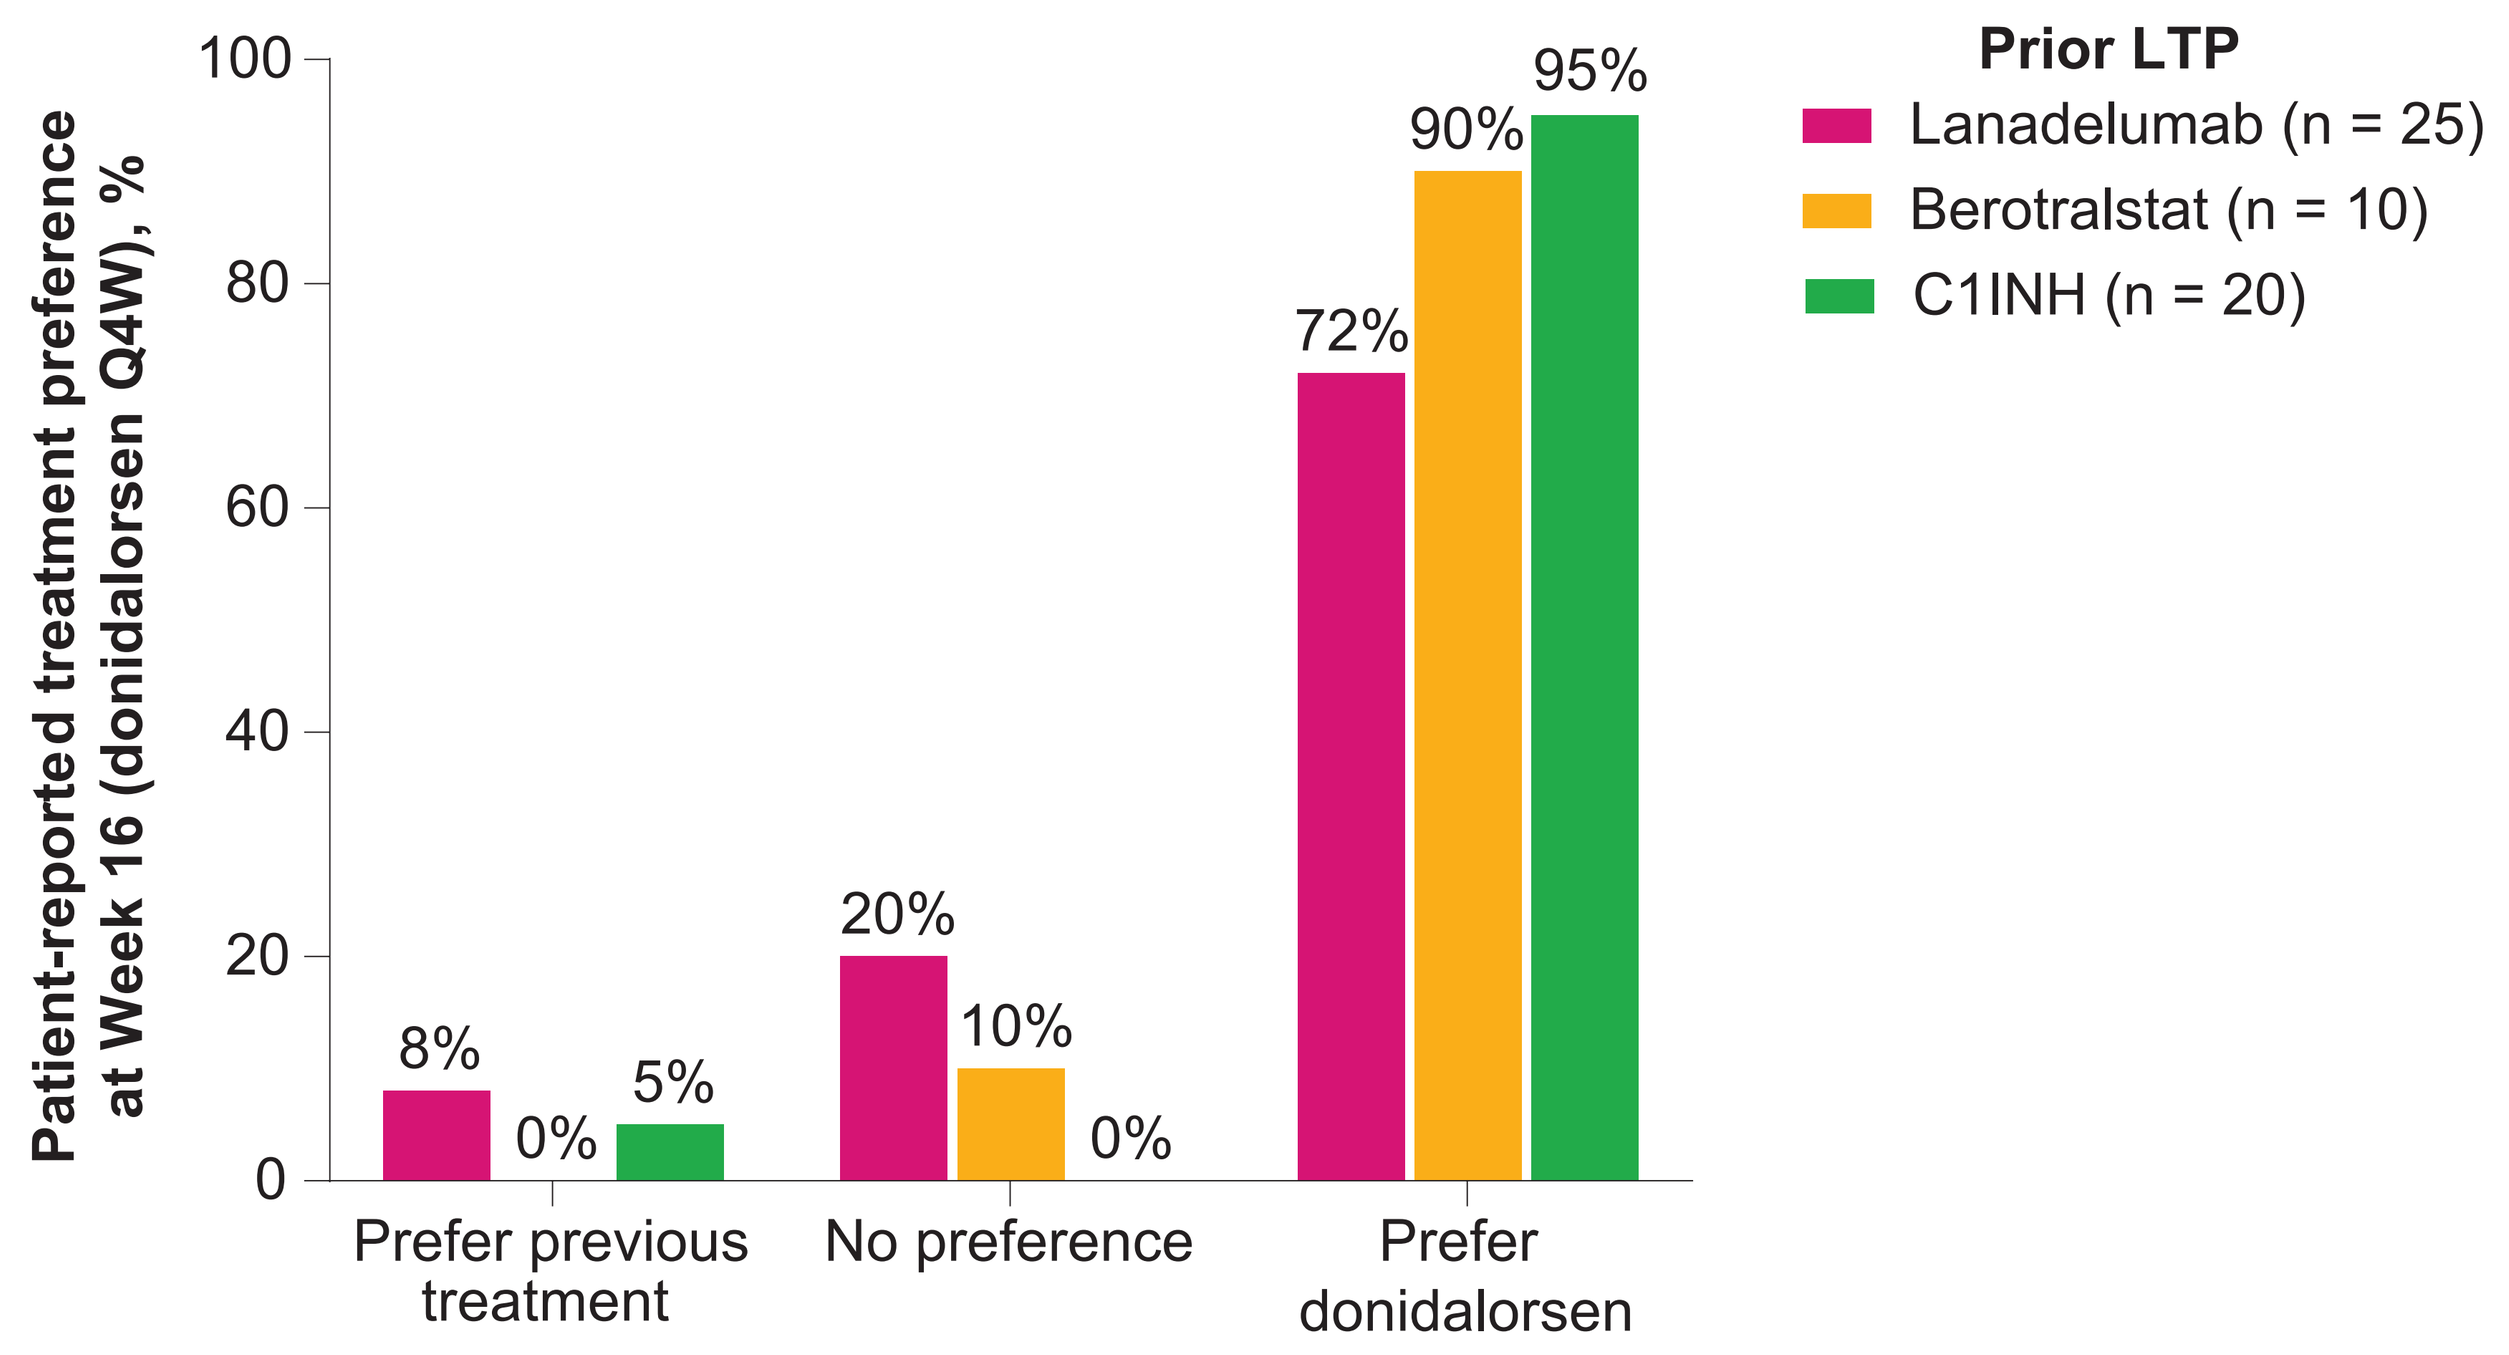

Patient-reported treatment preference at Week 16 (donidalorsen Q4W), %
0
20
40
60
80
100
8%
0%
5%
20%
10%
0%
72%
90%
95%
Prefer previous treatment
No preference
Prefer donidalorsen
Prior LTP
Lanadelumab (n = 25)
Berotralstat (n = 10)
C1INH (n = 20)

C1INH, C1 inhibitor; LTP, long-term prophylactic medication; Q4W, once every 4 weeks.

- Most patients preferred treatment with donidalorsen, regardless of their previous treatment
 - Overall, 84% of patients preferred donidalorsen, with 66% expressing a strong preference
- Patients most frequently selected the following reasons for their treatment preference:
 - **Lanadelumab:** Less injection-site pain or reactions with donidalorsen
 - **Berotralstat:** Better disease control with donidalorsen
 - **C1INH:** Less time for administration with donidalorsen

Figure 4. Global Treatment Satisfaction at Baseline and Week 16

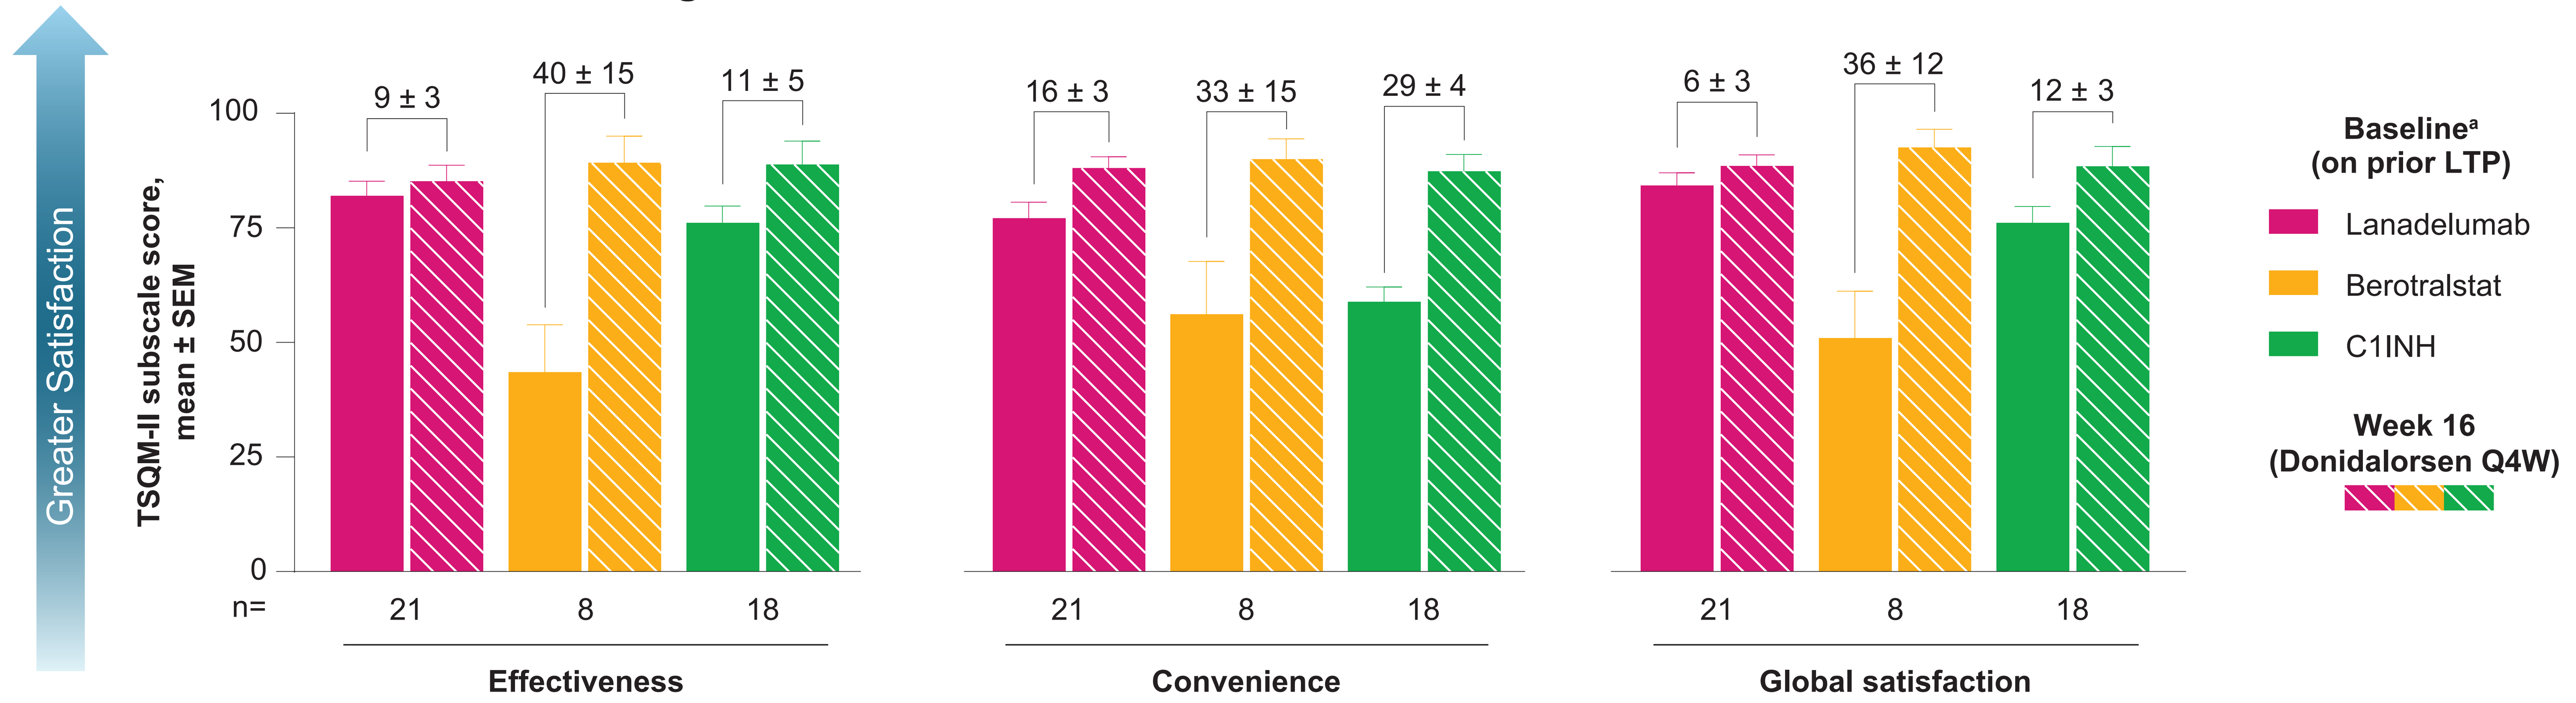

Greater Satisfaction
TSQM-II subscale score, mean ± SEM
0
25
50
75
100
9 ± 3
40 ± 15
11 ± 5
16 ± 3
33 ± 15
29 ± 4
6 ± 3
36 ± 12
12 ± 3
n=
21
8
18
21
8
18
21
8
18
Effectiveness
Convenience
Global satisfaction
Baseline[a] (on prior LTP)
Lanadelumab
Berotralstat
C1INH
Week 16 (Donidalorsen Q4W)

[a]Baseline is defined as the last measurement on or prior to day 1.
C1INH, C1 inhibitor; LTP, long-term prophylactic medication; Q4W, once every 4 weeks; SEM, standard error of the mean; TSQM-II, Treatment Satisfaction Questionnaire for Medication, version 2.

- TSQM-II scores for effectiveness, convenience, and global treatment satisfaction increased from baseline to Week 16 of donidalorsen treatment
- The side effect subscale of the TSQM-II was not reported as there were too few patients reporting side effects (≤5) at Week 16 to produce a reliable estimate

Conclusions

Safety and Tolerability

- Donidalorsen had an acceptable safety profile
- All injection-site reactions were mild or moderate in severity

Injection-Site Pain

- Patients reported improved injection-site pain over 16 weeks of donidalorsen treatment compared with the baseline assessment on their previous injectable LTP, particularly among those previously treated with lanadelumab

Treatment Satisfaction

- Patients reported improved convenience, effectiveness, and global satisfaction scores on the TSQM-II from their pre-switch assessment at baseline to Week 16 of treatment with donidalorsen 80 mg Q4W
 - **Prior lanadelumab:** largest improvement reported in convenience subscale
 - **Prior berotralstat:** improvements in all subscales
 - **Prior C1INH:** largest improvement reported in convenience subscale
- Most patients strongly preferred donidalorsen over their previous LTP

ACKNOWLEDGMENTS

We thank the patients and their families, the trial site coordinators, and the OASISplus team of investigators. We also thank Tracy Reigle, of Ionis, for graphic art assistance with the figures, and Nayna Sanathara, PhD, of Ionis, for editorial review. Medical writing and editorial assistance were provided by Red Nucleus and funded by Ionis Pharmaceuticals, Inc. This study was funded by Ionis Pharmaceuticals, Inc.

DISCLOSURES

MAR has received research grants from BioCryst, BioMarin Pharmaceutical, CSL Behring, Ionis, KalVista, Pharvaris, and Takeda; consulted for Astria, BioCryst, BioMarin Pharmaceutical, Celldex, CSL Behring, Cycle Pharma, Grifols, Intellia Therapeutics, Ionis, KalVista, Pfizer, Pharming, Pharvaris, Sanofi-Regeneron, and Takeda; and provided speaker presentations for CSL Behring, Grifols, Pharming, and Takeda. **JAB** has received research support from BioCryst, BioMarin Pharmaceutical, CSL Behring, Ionis, KalVista, Pharming, and Takeda; has provided speaker presentations for CSL Behring, Pharming, and Takeda; consulted for BioCryst, BioMarin Pharmaceutical, CSL Behring, Fresenius Kabi, Ionis, KalVista, Pharming, and Takeda; and is a member of the hereditary angioedema medical advisory board. **RT** has received grants or research support from Astria, BioCryst, CSL Behring, Ionis, KalVista, Pharvaris, and Takeda; is a speaker for AstraZeneca, BioCryst, CSL Behring, Grifols, GSK, Pharming, Sanofi-Regeneron, and Takeda; and has served as a consultant for BioCryst, CSL Behring, KalVista, Pharming, and Takeda. **AY**, **LB**, **KBN**, **ST**, and **TL** are employees of Ionis and hold shares and/or options in Ionis. **DMC** has received speaker or consultancy fees from Astria, BioCryst, CSL Behring, Intellia, Ionis, KalVista, Pharvaris, and Takeda.

REFERENCES

1. Riedl MA, et al. *J Allergy Clin Immunol Pract.* 2024;12(4):911-8.
2. Raasch J, et al. *World Allergy Organ J.* 2023;16(6):100792.
3. Busse P, et al. *J Allergy Clin Immunol Pract.* 2022;10(3):716-22.
4. Riedl MA, et al. *N Engl J Med.* 2024;391(1):21-31.
5. Atkinson MJ, et al. *Value Health.* 2005;8 Suppl 1:S9-S24.

American Academy of Allergy, Asthma, and Immunology/World Allergy Organization Joint Congress • San Diego, CA, USA • February 28–March 3, 2025 • Poster 599

d.m.cohn@amsterdamumc.nl