Poster 601

Hereditary Angioedema Disease Control After Switching to Donidalorsen From Prior Long-Term Prophylaxis: Results From the OASISplus Open-Label Extension Study

Marc A Riedl[1], Jonathan A Bernstein[2], Laura Bordone[3], Raffi Tachdjian[4], Kenneth B Newman[3], Sabrina Treadwell[3], Tao Lin[3], Aaron Yarlas[3], Danny M Cohn[5]

[1]Division of Allergy and Immunology, University of California San Diego School of Medicine, La Jolla, CA, USA; [2]Division of Rheumatology, Allergy, and Immunology, Department of Internal Medicine, University of Cincinnati College of Medicine, Cincinnati, OH, USA; [3]Ionis, Carlsbad, CA, USA; [4]Division of Allergy, Immunology, and Rheumatology, University of California Los Angeles, Los Angeles, CA, USA; [5]Department of Vascular Medicine, Amsterdam Cardiovascular Sciences, Amsterdam UMC, University of Amsterdam, Amsterdam, the Netherlands

Background

- Hereditary angioedema (HAE) is a rare disease characterized by frequently severe, potentially life-threatening attacks of tissue swelling[1,2]
- HAE is most often caused by C1 inhibitor (C1INH) deficiency (HAE-C1INH-Type1) or dysfunction (HAE-C1INH-Type2) which leads to dysregulation of the kallikrein-kinin system[3]
- Donidalorsen is an investigational RNA-targeted antisense oligonucleotide that specifically reduces prekallikrein production in the liver[4]
- In the phase 3 OASIS-HAE study (NCT05139810), donidalorsen 80 mg subcutaneously (SC) once every 4 weeks (Q4W) significantly reduced HAE attack rate and improved patient-reported disease control[4]
- In the ongoing OASISplus open-label extension (OLE) study (NCT05392114), patients who switched from a previous long-term prophylactic medication (LTP) to donidalorsen (Switch cohort) experienced lower HAE attack rates and improved quality of life as measured by the Angioedema Quality of Life Questionnaire[5]
- Here, we report additional disease control results for patients in the Switch cohort at Week 16 of the OLE from a February 2024 data cutoff

Study Design

Figure 1. Study Design

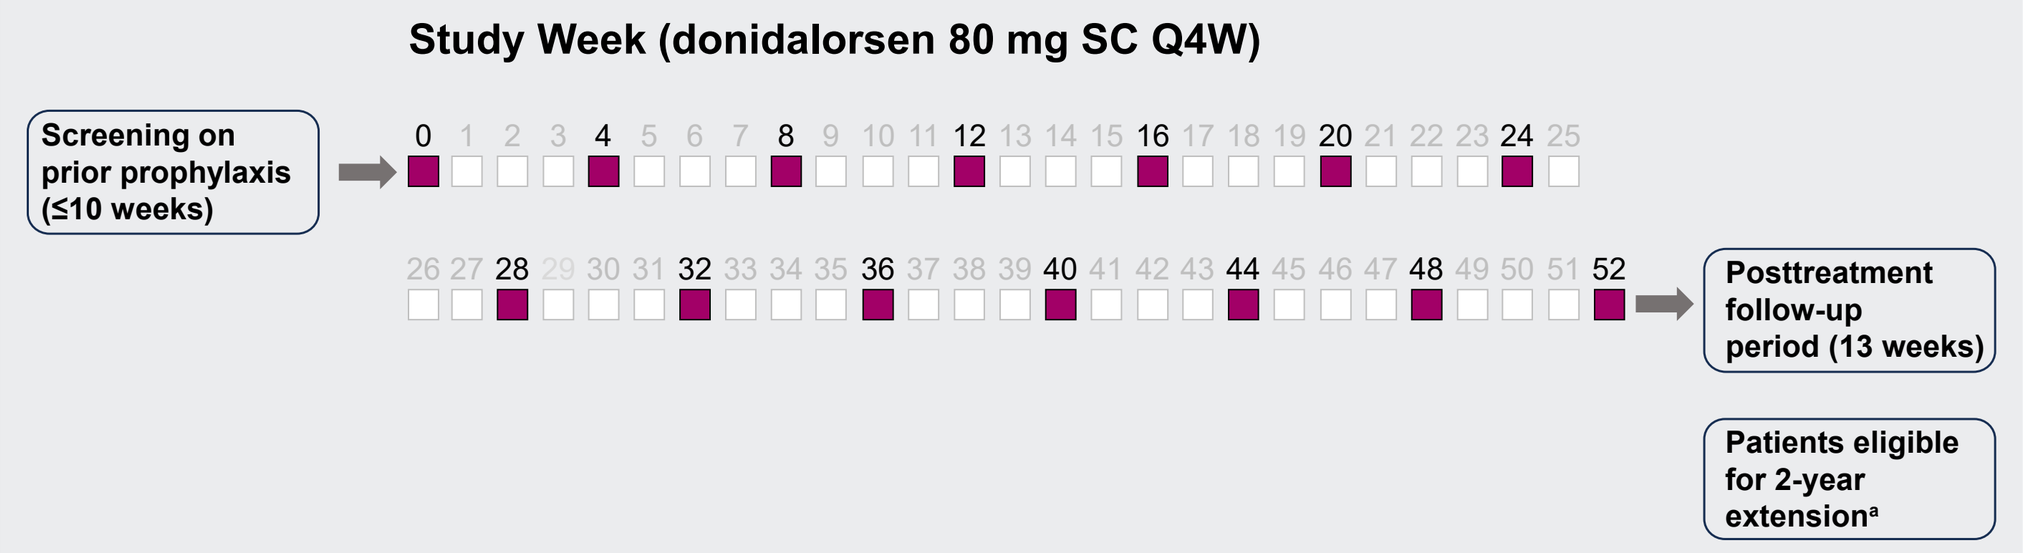
Study Week (donidalorsen 80 mg SC Q4W)
Screening on prior prophylaxis (≤10 weeks)
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25
26 27 28 29 30 31 32 33 34 35 36 37 38 39 40 41 42 43 44 45 46 47 48 49 50 51 52
Posttreatment follow-up period (13 weeks)
Patients eligible for 2-year extension[a]

[a]Patients could switch to Q8W dosing after Year 1.
Q4W, once every 4 weeks; Q8W, once every 8 weeks; SC, subcutaneously.

- Patients were ≥12 years of age with HAE-C1INH-Type1 or HAE-C1INH-Type2 on stable doses of lanadelumab, berotralstat, or C1INH for ≥12 weeks
- All patients switched to donidalorsen 80 mg SC Q4W with no washout period
- Patients received the last dose of their prior LTP based on a prespecified algorithm as follows:
 - Lanadelumab: 14 ± 3 days before day 1
 - Berotralstat: 14 ± 3 days after day 1
 - C1INH: 14 ± 3 days after day 1
- Endpoints reported here include:
 - Incidence and severity of treatment-emergent adverse events (TEAEs; primary endpoint)
 - Time-normalized rate of investigator-confirmed HAE attacks per month from baseline to Week 16 (prespecified exploratory endpoint)
 - Angioedema Control Test (AECT; prespecified exploratory endpoint)
 - Scores range from 0–16; higher scores indicate better disease control[6]
 - Scores ≥10 indicate well-controlled disease[7]
 - Minimum clinically important difference (MCID): 3-point increase[8]
- Data are shown from an interim February 2024 data cutoff

Results

Table 1. Patient Disposition and Demographics

	Patients switching to donidalorsen 80 mg Q4W from:			
	Lanadelumab	Berotralstat	C1INH	Total
Patients enrolled, n	32	11	22	65
Patients dosed, n	31	11	22	64
Completed Week 16 of treatment, n (%)	28 (88)	10 (91)	20 (91)	58 (89)
Early termination, n (%)				
Lack of efficacy	3 (9)	0	1 (5)	4 (6)
Adverse event	1[a] (3)	0	0	1[a] (2)
Lost to follow-up	1 (3)	0	0	1 (2)
Voluntary withdrawal	0	0	1 (5)	1 (2)
Other (not dosed)	1 (3)	0	0	1 (2)
Age, years, mean (SD)	40 (14)	46 (11)	41 (17)	42 (15)
Age, years, n (%)				
12–17 years of age	1 (3)	0	3 (14)	4 (6)
≥18 years of age	30 (97)	11 (100)	19 (86)	60 (94)
Sex, n (%)				
Male	17 (55)	3 (27)	6 (27)	26 (41)
Female	14 (45)	8 (73)	16 (73)	38 (59)
Race,[b] n (%)				
White	26 (84)	11 (100)	20 (91)	57 (89)
Multiple or other[c]	5 (16)	0	2 (9)	7 (11)

[a]One patient discontinued due to a series of cardiac failure and renal disorder TEAEs, including serious TEAEs of heart failure exacerbation and renal infarct, that were not related to the study drug. [b]Race was self-reported by patients during screening. [c]Includes American Indian or Alaska Native, Asian, Black or African American, Native Hawaiian or Other Pacific Islander, multiple races, and other races
C1INH, C1 inhibitor; Q4W, once every 4 weeks; SD, standard deviation.

- As of February 28, 2024, 58 of 64 dosed patients (89%) had completed Week 16 of treatment
- Mean exposure to donidalorsen was 263 days

Table 2. Overview of Treatment-Emergent Adverse Events (Weeks 0–16)

	Donidalorsen Q4W (N = 64)
Any TEAE,[a] n (%)	45 (70)
Related to study drug	17 (27)
Leading to discontinuation	1[b] (2)
Any serious TEAE, n (%)	1[b] (2)
Related to study drug	0
Severity of TEAEs, n (%)	
Mild	17 (27)
Moderate	25 (39)
Severe	3 (5)
Severity of TEAEs related to study drug, n (%)	
Mild	12 (19)
Moderate	4 (6)
Severe	1[c] (2)

[a]TEAE is defined as any adverse event starting or worsening on or after the first dose of donidalorsen. [b]One patient discontinued due to a series of cardiac failure and renal disorder TEAEs, including serious TEAEs of heart failure exacerbation and renal infarct, that were not related to the study drug. [c]The TEAE was a headache, and no action was taken with the study drug.
Q4W, once every 4 weeks; TEAE, treatment-emergent adverse event.

- Most TEAEs were mild or moderate in severity

Figure 2. HAE Attack Rate (Weeks 0–16)

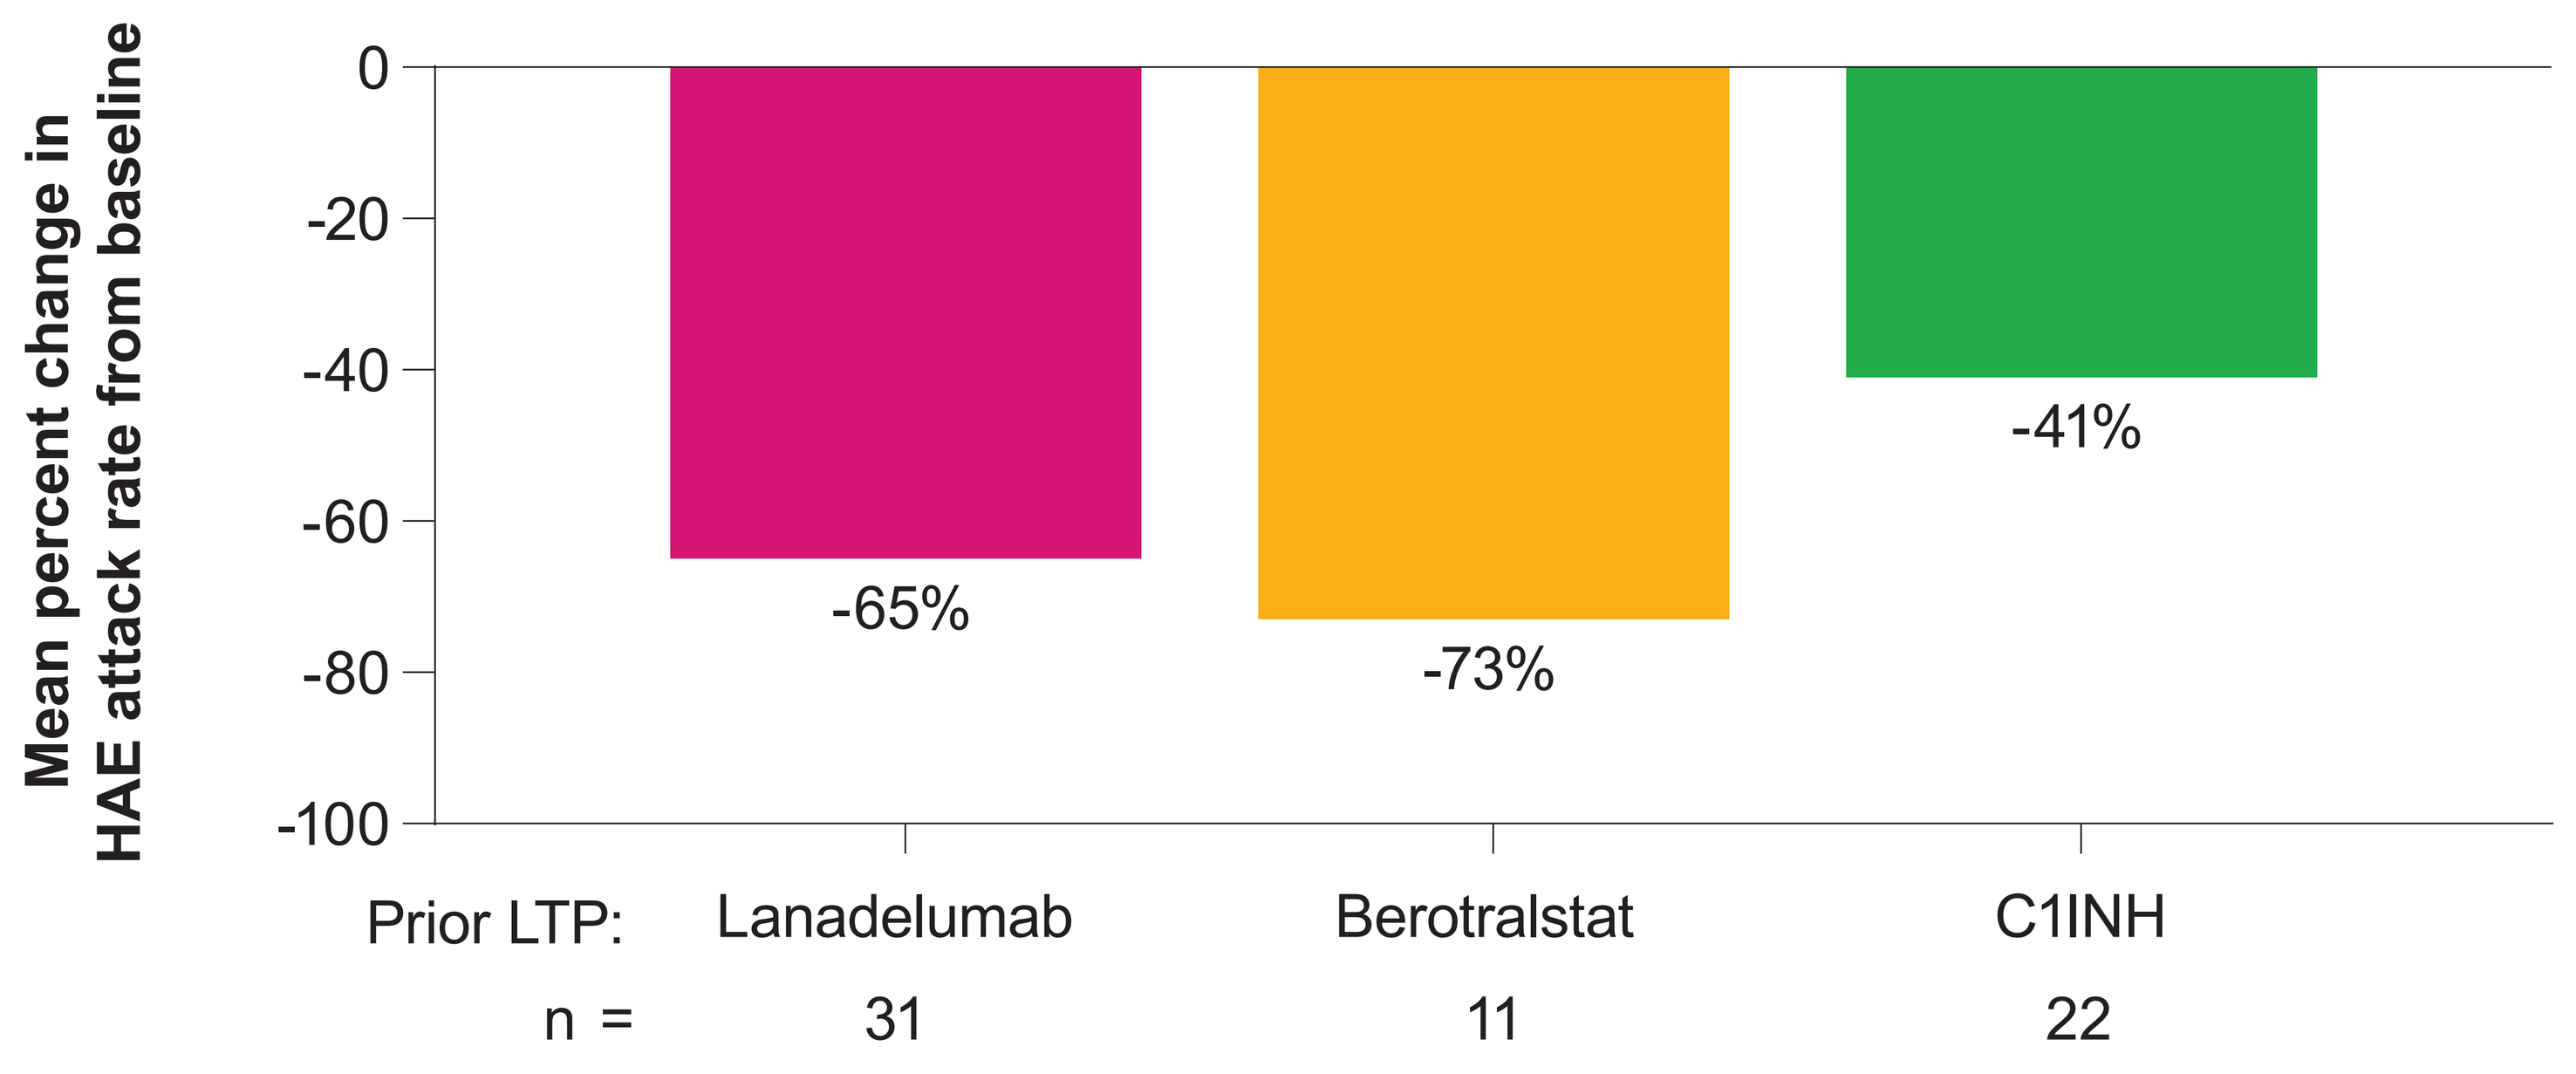
Mean percent change in HAE attack rate from baseline
0
-20
-40
-60
-80
-100
-65%
-73%
-41%
Prior LTP: Lanadelumab Berotralstat C1INH
n = 31 11 22

	Lanadelumab	Berotralstat	C1INH
Baseline[a] (on prior LTP), mean ± SD =	0.69 ± 1.09	1.80 ± 1.92	0.61 ± 0.96
Week 16 (donidalorsen Q4W), mean ± SD =	0.24 ± 0.33	0.48 ± 0.53	0.36 ± 0.69

[a]Baseline HAE attack rate during the screening period for the Switch cohort.
C1INH, C1 inhibitor; HAE, hereditary angioedema; LTP, long-term prophylactic medication; Q4W, once every 4 weeks; SD, standard deviation.

- There was an overall 62% mean decrease from baseline in HAE attack rate through Week 16

Figure 3. HAE Attack Rate in Patients With Poorly Controlled Disease at Baseline (Weeks 0–16)

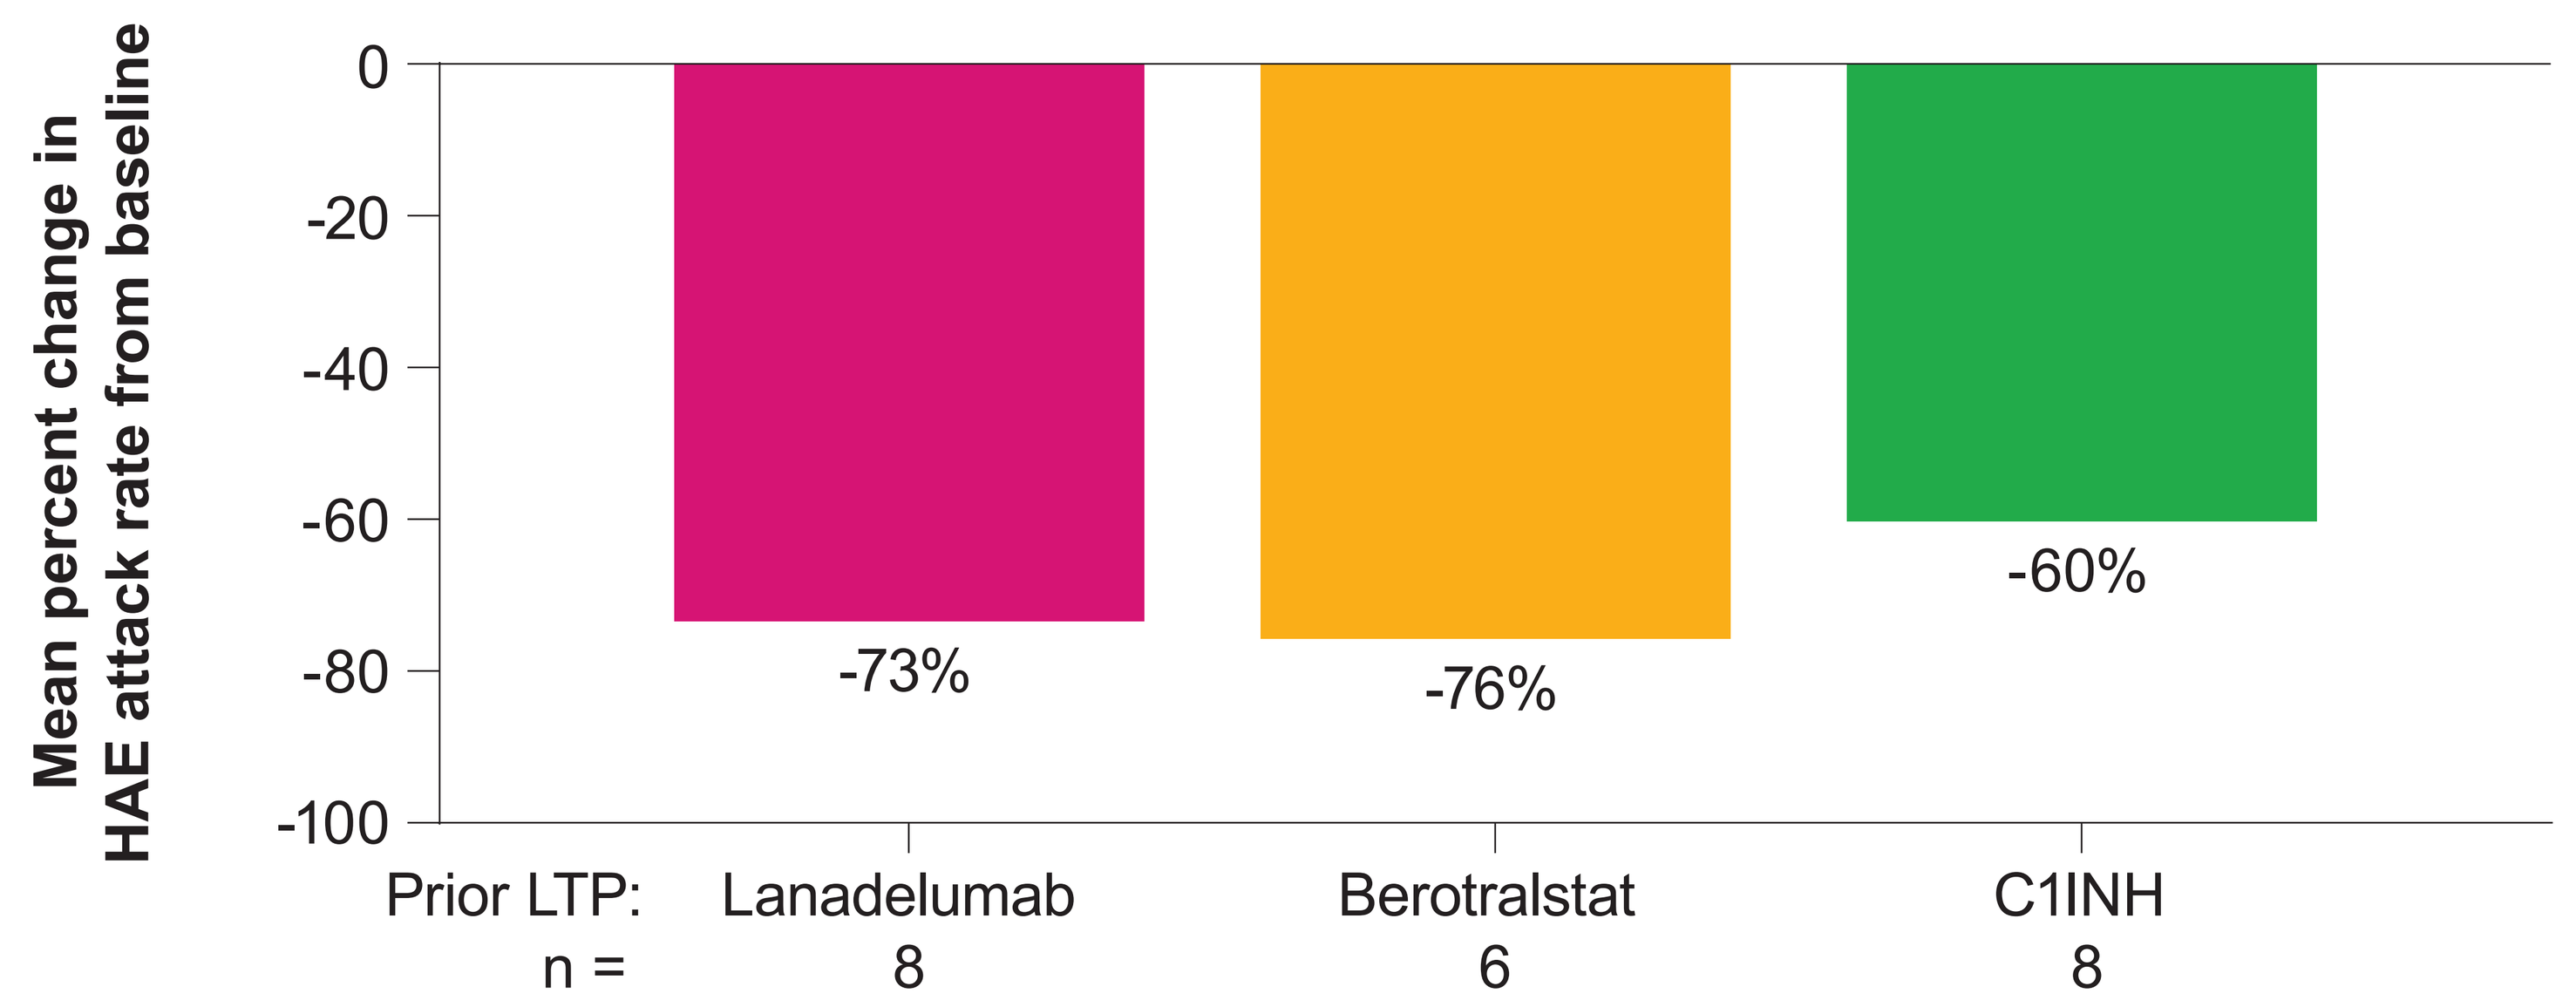
Mean percent change in HAE attack rate from baseline
0
-20
-40
-60
-80
-100
-73%
-76%
-60%
Prior LTP: Lanadelumab Berotralstat C1INH
n = 8 6 8

	Lanadelumab	Berotralstat	C1INH
Baseline[a] (on prior LTP), mean ± SD =	1.68 ± 1.34	2.54 ± 2.12	1.19 ± 1.22
Week 16 (donidalorsen Q4W), mean ± SD =	0.45 ± 0.37	0.62 ± 0.56	0.47 ± 0.82

[a]Baseline HAE attack rate during the screening period for the Switch cohort.
C1INH, C1 inhibitor; HAE, hereditary angioedema; LTP, long-term prophylactic medication; Q4W, once every 4 weeks; SD, standard deviation.

- HAE attack rate decreased by an overall mean of 71% in patients who reported poorly controlled disease (AECT <10)[7] at baseline

Figure 4. Mean AECT Total Scores (Week 16)

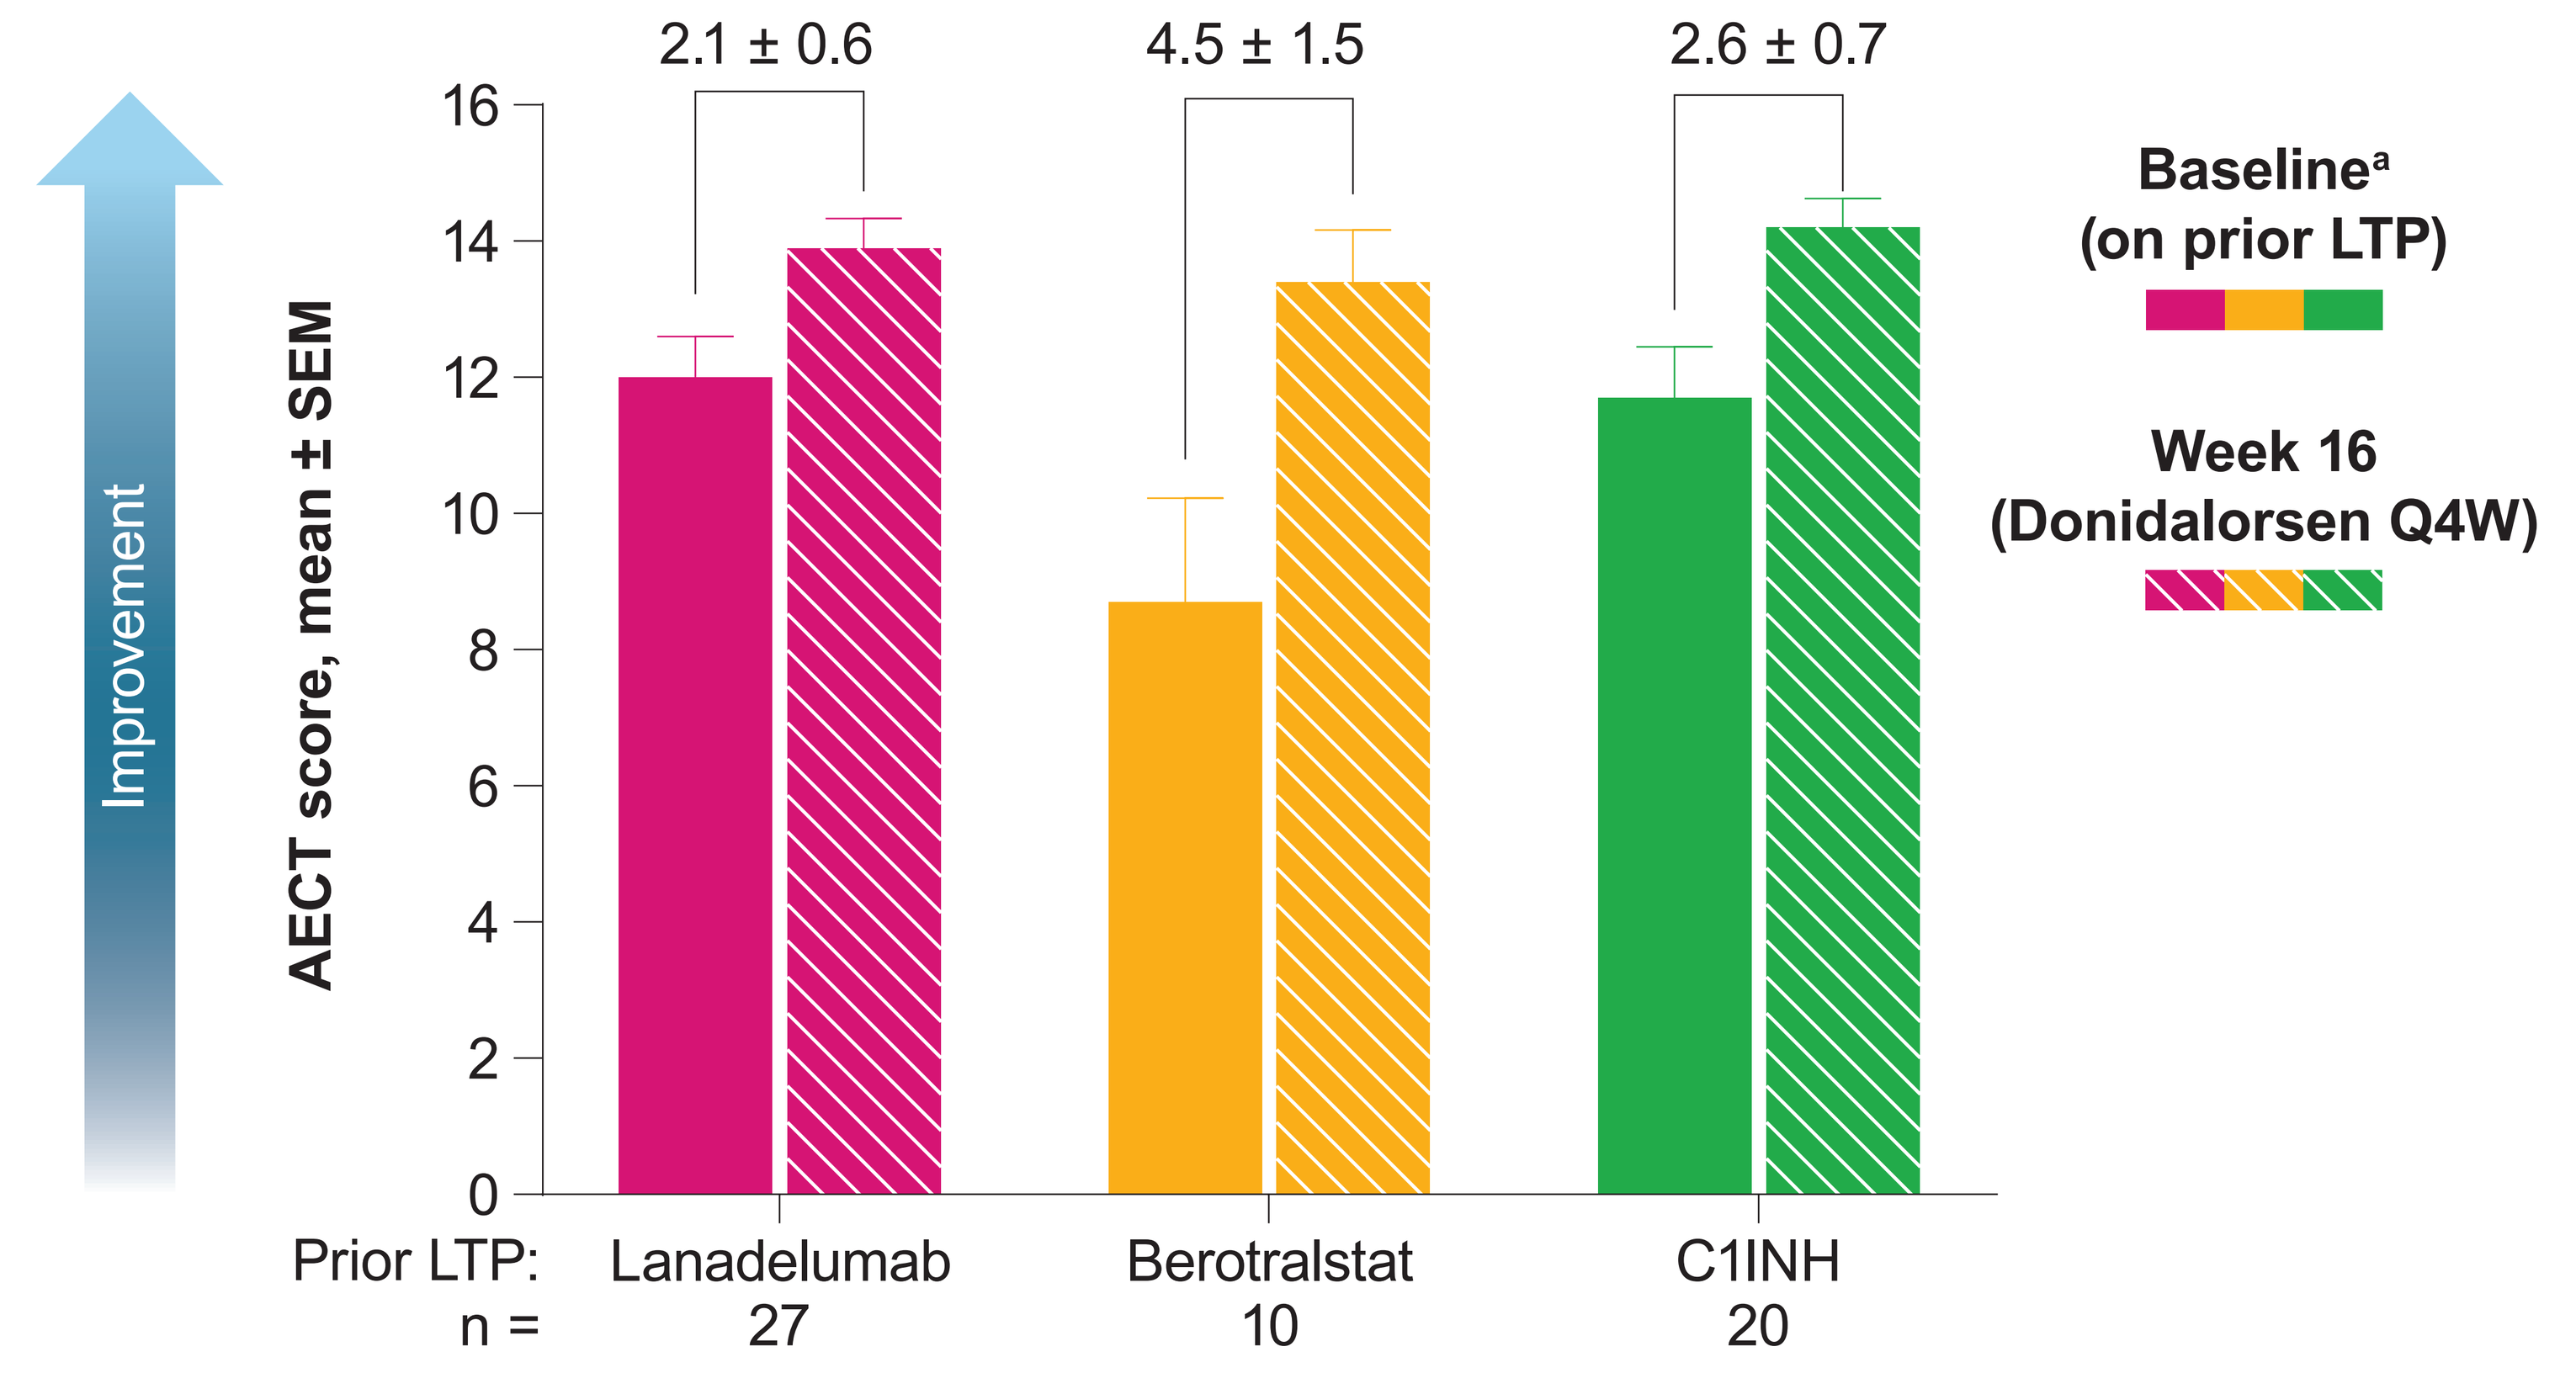
Improvement
AECT score, mean ± SEM
16
14
12
10
8
6
4
2
0
2.1 ± 0.6
4.5 ± 1.5
2.6 ± 0.7
Prior LTP: Lanadelumab Berotralstat C1INH
n = 27 10 20
Baseline[a] (on prior LTP)
Week 16 (Donidalorsen Q4W)

[a]The last measurement on or before day 1.
AECT, Angioedema Control Test; C1INH, C1 inhibitor; LTP, long-term prophylactic medication; Q4W, once every 4 weeks; SEM, standard error of the mean.

- Overall, mean (standard deviation) AECT total scores increased from 11.3 (3.9) at baseline to 13.9 (2.2) with donidalorsen 80 mg SC Q4W at Week 16
- The mean increase in AECT score exceeded the MCID (3-point increase)[8] in patients who switched to donidalorsen from berotralstat

Figure 5. Percentage of Patients With Well-Controlled Disease (Week 16)

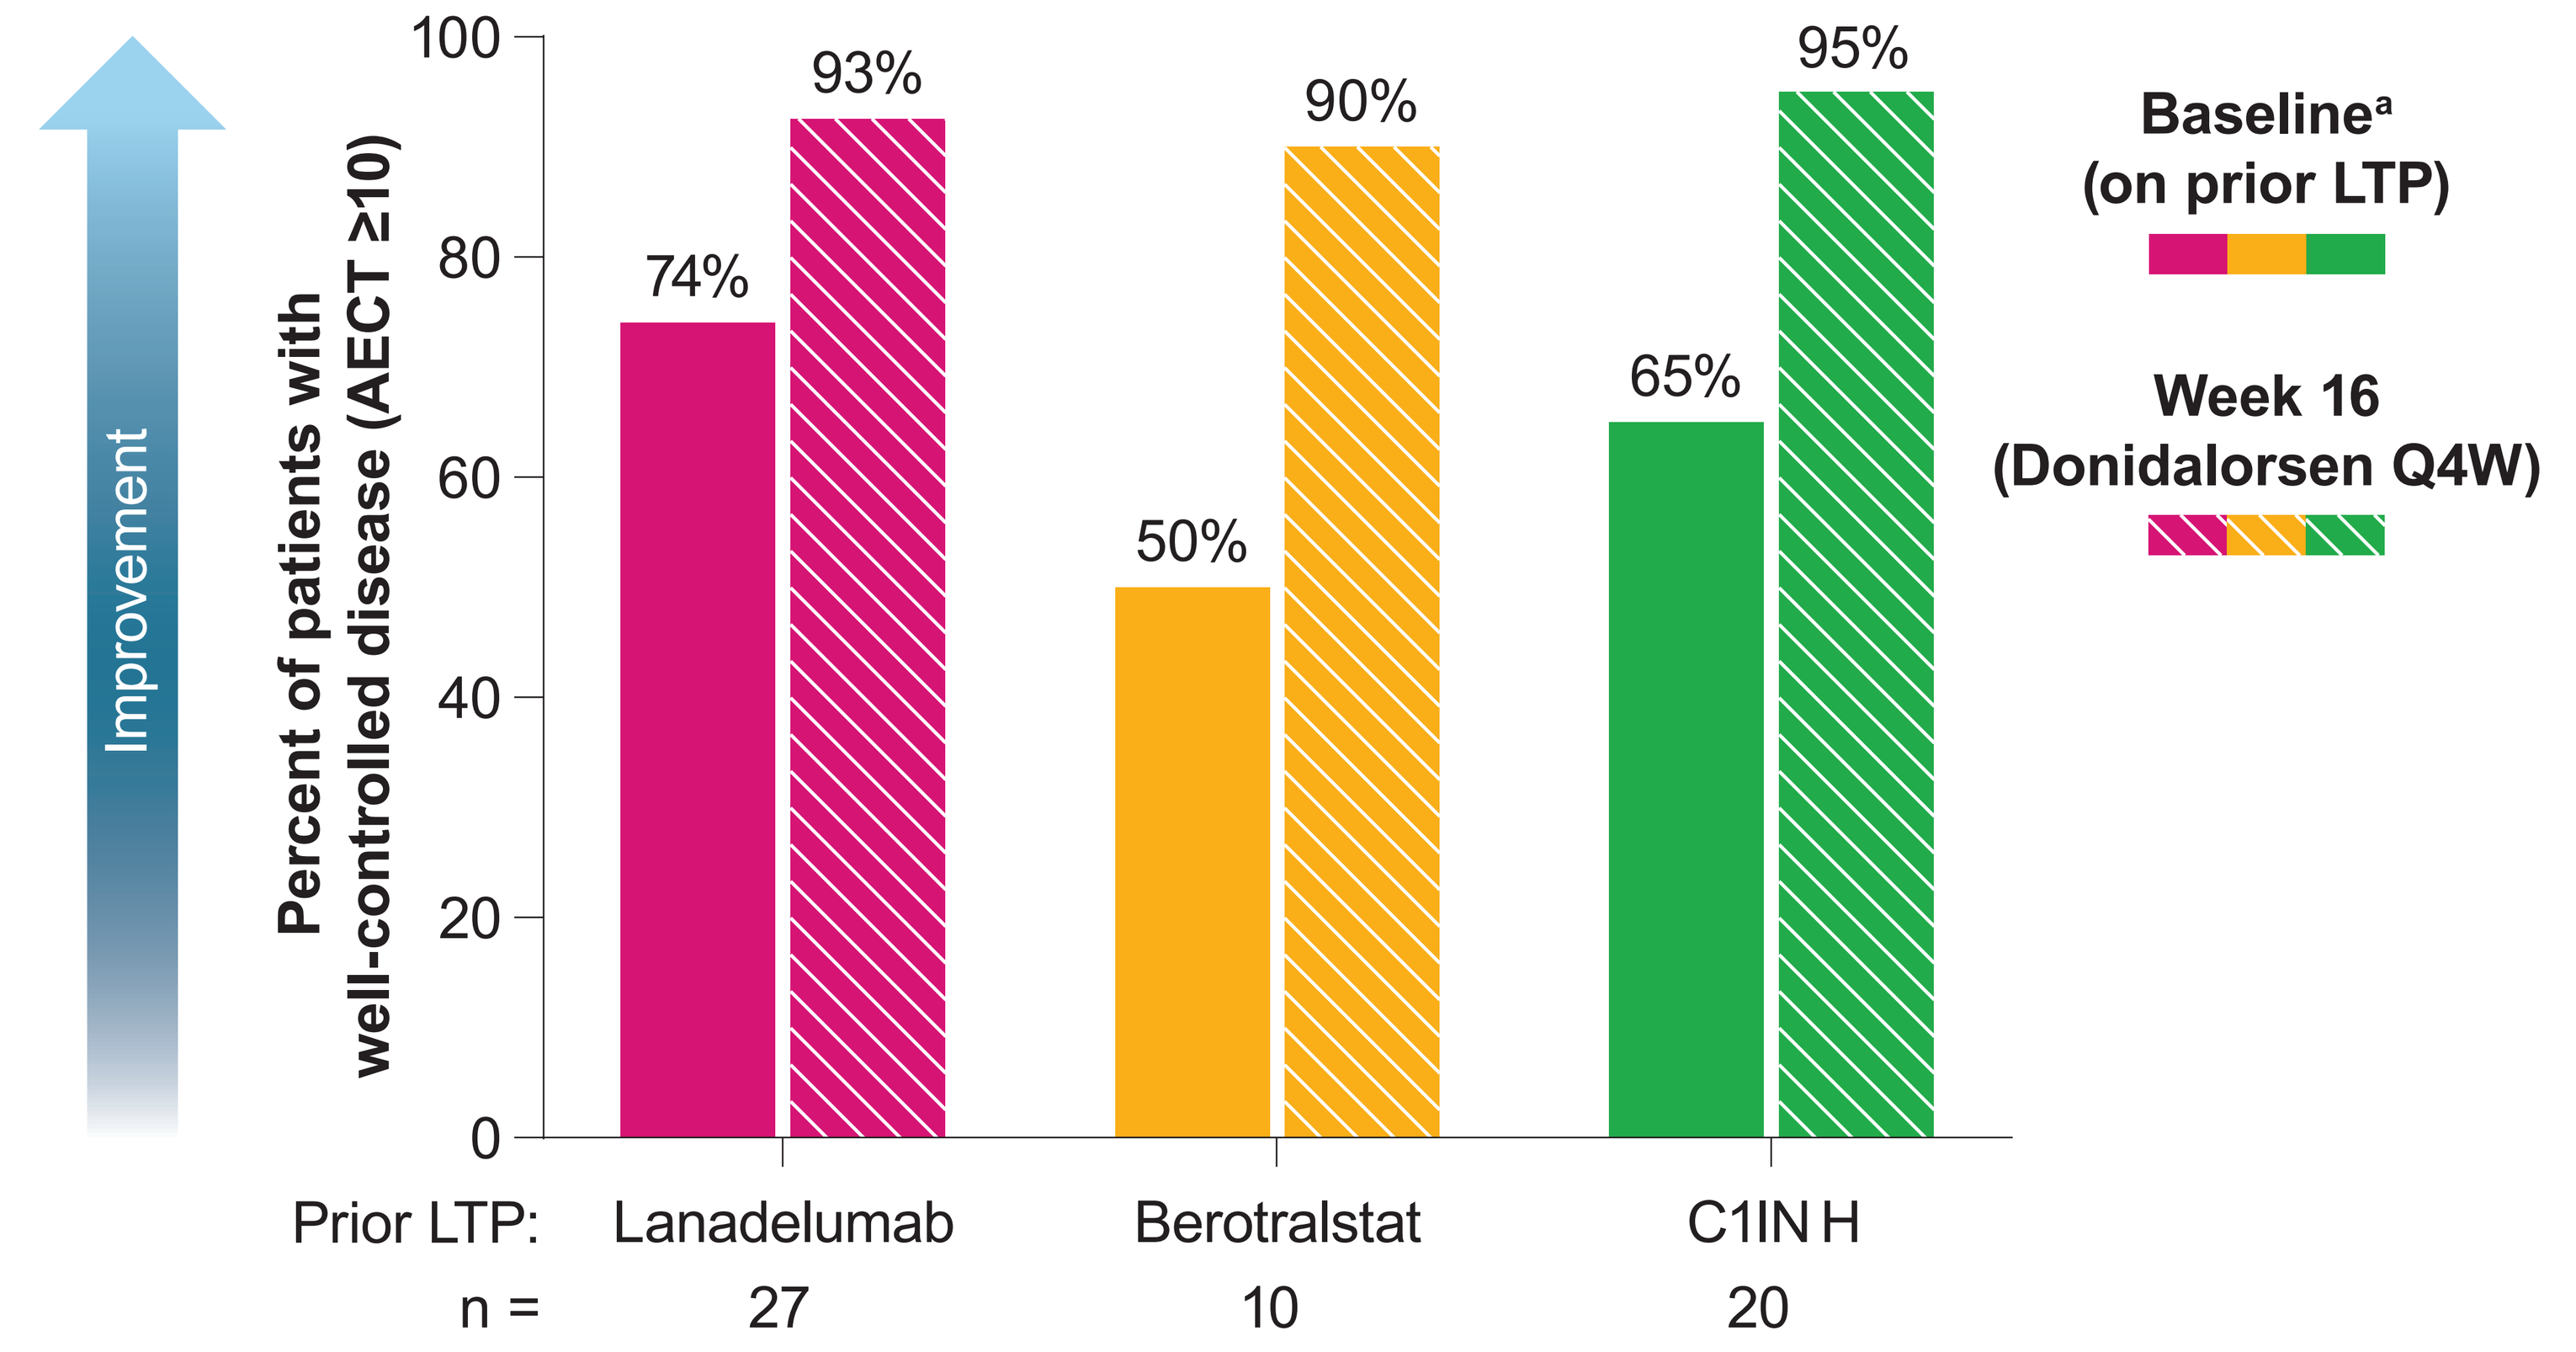
Improvement
Percent of patients with well-controlled disease (AECT ≥10)
100
80
60
40
20
0
74%
93%
50%
90%
65%
95%
Prior LTP: Lanadelumab Berotralstat C1INH
n = 27 10 20
Baseline[a] (on prior LTP)
Week 16 (Donidalorsen Q4W)

[a]The last measurement on or before day 1.
AECT, Angioedema Control Test; C1INH, C1 inhibitor; LTP, long-term prophylactic medication; Q4W, once every 4 weeks.

- The overall percentage of patients reporting well-controlled disease (AECT ≥10)[7] increased from 67% at baseline to 93% with donidalorsen 80 mg SC Q4W at Week 16
- At Week 16, ≥90% of patients in each group reported well-controlled disease

Conclusions

Safety

- Donidalorsen had an acceptable safety and tolerability profile in patients switching from prior long-term prophylactic medications

HAE Attack Rate

- Through Week 16 of donidalorsen treatment, HAE attack rates decreased by an overall mean of 62% compared with prior long-term prophylactic medications and by 71% in patients with poorly controlled disease at baseline

Disease Control

- Patients reported improvements in disease control after switching to donidalorsen
- By Week 16, the proportion of patients reporting well-controlled disease was ≥90% in all groups

ACKNOWLEDGMENTS

We thank the patients who participated in this trial, the trial site coordinators, and the OASISplus team of investigators. We also thank Tracy Reigle, of Ionis, for graphic art assistance with the figures, and Nayna Sanathara, PhD, of Ionis, for editorial review. Medical writing and editorial assistance were provided by Jake Wilmot, PhD, of Red Nucleus, and funded by Ionis Pharmaceuticals, Inc. This study was funded by Ionis Pharmaceuticals, Inc.

DISCLOSURES

MAR has received research grants from BioCryst, BioMarin Pharmaceutical, CSL Behring, Ionis, KalVista, Pharvaris, and Takeda; consulted for Astria, BioCryst, BioMarin Pharmaceutical, Celldex, CSL Behring, Cycle Pharma, Grifols, Intellia Therapeutics, Ionis, KalVista, Pfizer, Pharming, Pharvaris, Sanofi-Regeneron, and Takeda; and provided speaker presentations for CSL Behring, Grifols, Pharming, and Takeda. **JAB** has received research support from BioCryst, BioMarin Pharmaceutical, CSL Behring, Ionis, KalVista, Pharming, and Takeda; has provided speaker presentations for CSL Behring, Pharming, and Takeda; consulted for BioCryst, BioMarin Pharmaceutical, CSL Behring, Fresenius Kabi, Ionis, KalVista, Pharming, and Takeda; and is a member of the hereditary angioedema medical advisory board. **LB, KBN, ST, TL,** and **AY** are employees of Ionis and hold shares and/or options in Ionis. **RT** has received grants or research support from Astria, BioCryst, CSL Behring, Ionis, KalVista, Pharvaris, and Takeda; is a speaker for AstraZeneca, BioCryst, CSL Behring, Grifols, GSK, Pharming, Sanofi-Regeneron, and Takeda; and has served as a consultant for BioCryst, CSL Behring, KalVista, Pharming, and Takeda. **DMC** has received speaker or consultancy fees from Astria, BioCryst, CSL Behring, Intellia, Ionis, KalVista, Pharvaris, and Takeda.

REFERENCES

1. Riedl MA, et al. *J Allergy Clin Immunol Pract*. 2024;12(4):911-8.
2. Raasch J, et al. *World Allergy Organ J*. 2023;16(6):100792.
3. Busse P, et al. *J Allergy Clin Immunol Pract*. 2022;10(3):716-22.
4. Riedl MA, et al. *N Engl J Med*. 2024;391(1):21-31.
5. Riedl MA, et al. Presented at EAACI. 2024.
6. Weller K, et al. *Allergy*. 2020;75(5):1165–1177.
7. Weller K, et al. *J Allergy Clin Immunol Pract*. 2020;8(6):2050-7 e4.
8. Fijen LM, et al. *Clin Transl Allergy*. 2023;13(9):e12295.

American Academy of Allergy, Asthma, and Immunology/World Allergy Organization Joint Congress • San Diego, CA, USA • February 28–March 3, 2025 • Poster 601

mriedl@health.ucsd.edu