Poster 654

Long-Term Analysis of the Phase 2 Open-Label Extension of Donidalorsen in Patients With Hereditary Angioedema

Michael Manning[1], Laura Bordone[2], Danny M. Cohn[3], Yiwen Deng[2], Sabrina Treadwell[2], Kenneth B. Newman[2], Marc A. Riedl[4], Raffi Tachdjian[5]

[1]Medical Research of Arizona, Scottsdale, AZ, USA; [2]Ionis, Carlsbad, CA, USA; [3]Department of Vascular Medicine, Amsterdam Cardiovascular Sciences, Amsterdam UMC, University of Amsterdam, Amsterdam, the Netherlands; [4]Division of Allergy and Immunology, University of California San Diego, La Jolla, CA, USA; [5]Division of Allergy, Immunology, and Rheumatology, University of California Los Angeles, Los Angeles, CA, USA

Background

- Hereditary angioedema (HAE) is a rare disease characterized by recurrent, potentially life-threatening attacks of tissue swelling[1,2]
- In patients with HAE, C1 inhibitor (C1INH) deficiency (HAE-C1INH-Type1) or dysfunction (HAE-C1INH-Type2) leads to dysregulation of the kallikrein-kinin system[3]
- Donidalorsen is an investigational RNA-targeted antisense oligonucleotide that specifically reduces prekallikrein production in the liver[4]
- Reduction of prekallikrein concentration stabilizes the kallikrein-kinin system in patients with HAE[5]
- In a phase 2 randomized clinical trial (NCT04030598), donidalorsen led to a 90% reduction in HAE attacks vs placebo[5]
- Here, we report the safety and efficacy of donidalorsen from an interim analysis from June 2024 of the open-label extension (OLE; NCT04307381) of the phase 2 randomized study in patients with HAE-C1INH-Type1 or HAE-C1INH-Type2

Study Design

Figure 1. Study Design

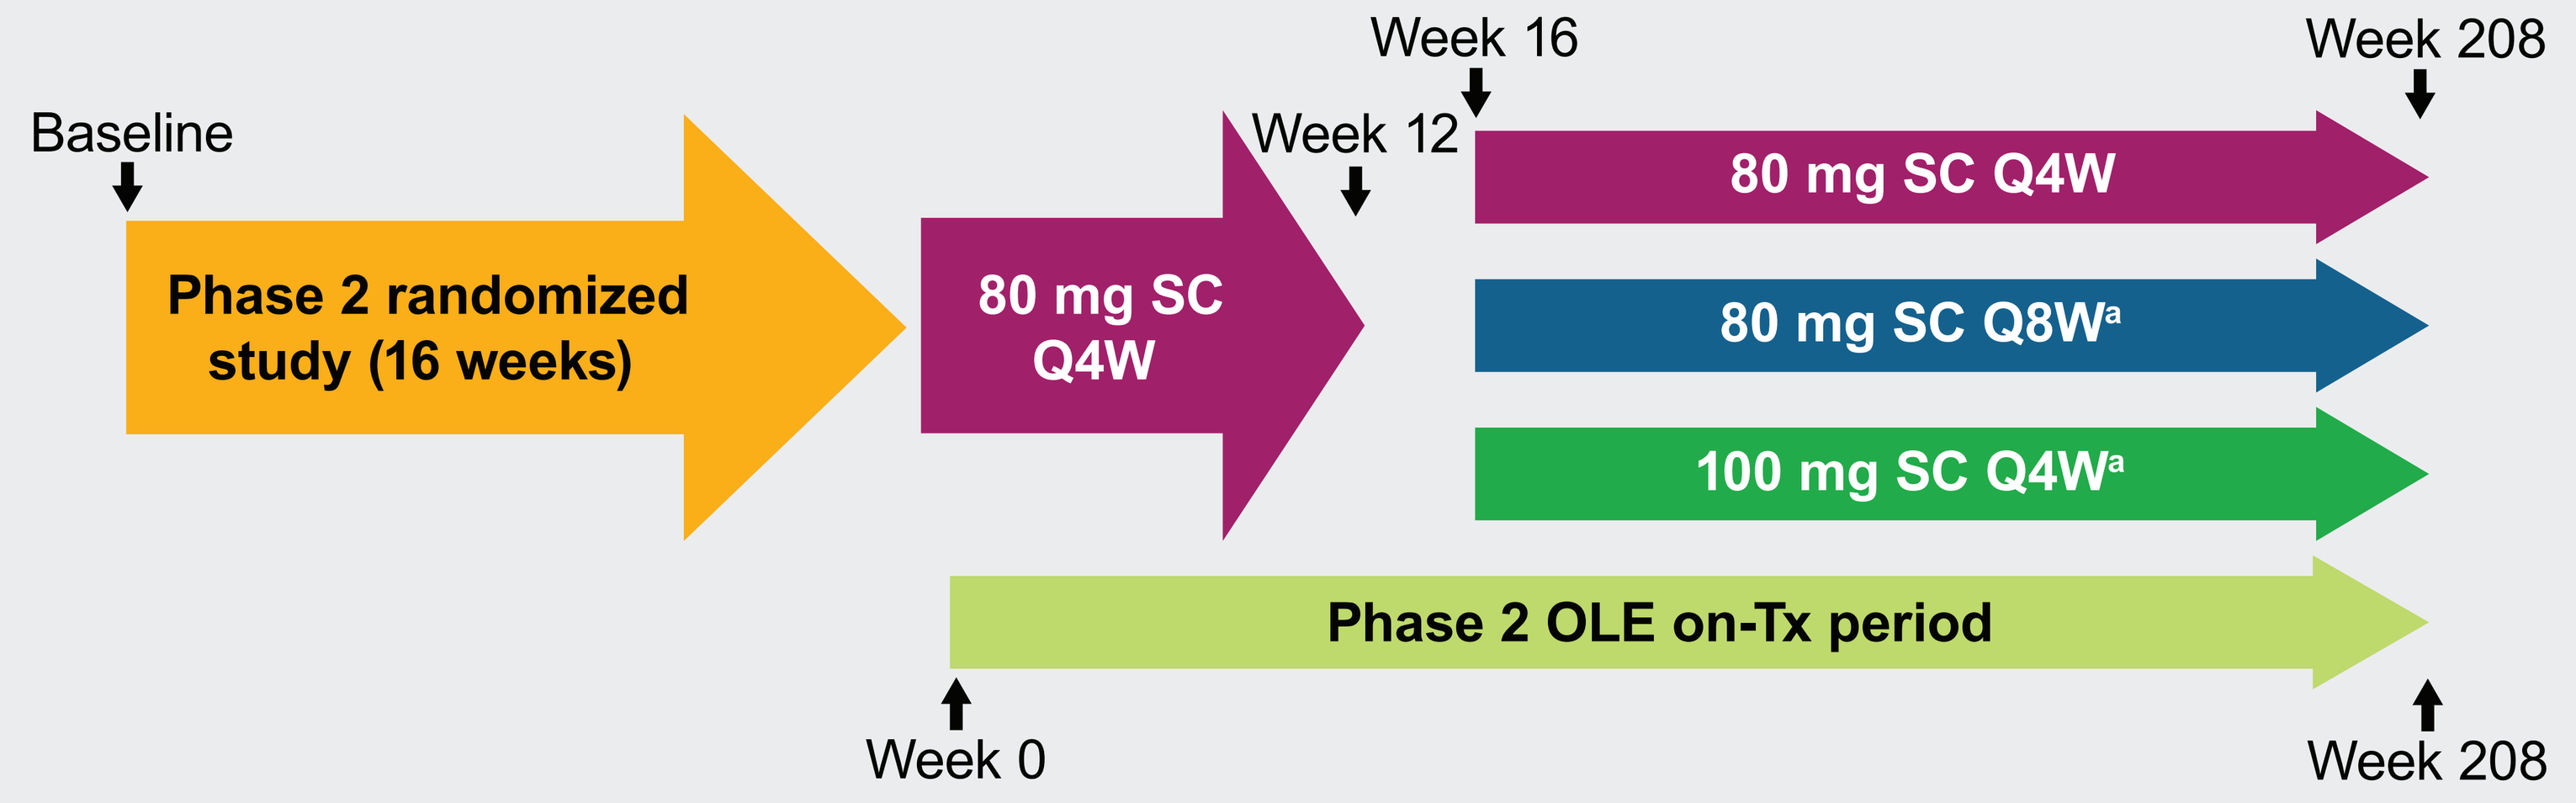
Baseline
Phase 2 randomized study (16 weeks)
80 mg SC Q4W
Week 12
Week 16
Week 208
80 mg SC Q4W
80 mg SC Q8W[a]
100 mg SC Q4W[a]
Phase 2 OLE on-Tx period
Week 0
Week 208

[a]Switch in dosing regimen per the principal investigator.
Weeks 0–16 were termed Weeks 1–17 in previous publications. Time points have been adjusted to start at Week 0 to align with the 4-week dosing schedule.
OLE, open-label extension; Q4W, once every 4 weeks; Q8W, once every 8 weeks; SC, subcutaneously; Tx, treatment.

- Eligible patients were ≥18 years of age and completed the phase 2 randomized study
- The treatment period consisted of fixed and flexible treatment periods:
 - In the fixed treatment period (Weeks 0–12), all patients received donidalorsen 80 mg subcutaneously (SC) once every 4 weeks (Q4W)
 - In the flexible treatment period (Weeks 16–208),
 - Patients could switch to 80 mg SC once every 8 weeks (Q8W) if they were attack free for ≥12 weeks
 - Patients who experienced HAE attacks in the prior 12 weeks could switch to 100 mg SC Q4W
- Endpoints included the following:
 - Incidence and severity of treatment-emergent adverse events (TEAEs)
 - Time-normalized HAE attacks (monthly)
 - Angioedema Quality of Life Questionnaire (AE-QoL) score at the prespecified Week 156 time point
- Endpoints summarize data from the treatment period compared with baseline from the phase 2 randomized study

Results

Figure 2. Patient Disposition

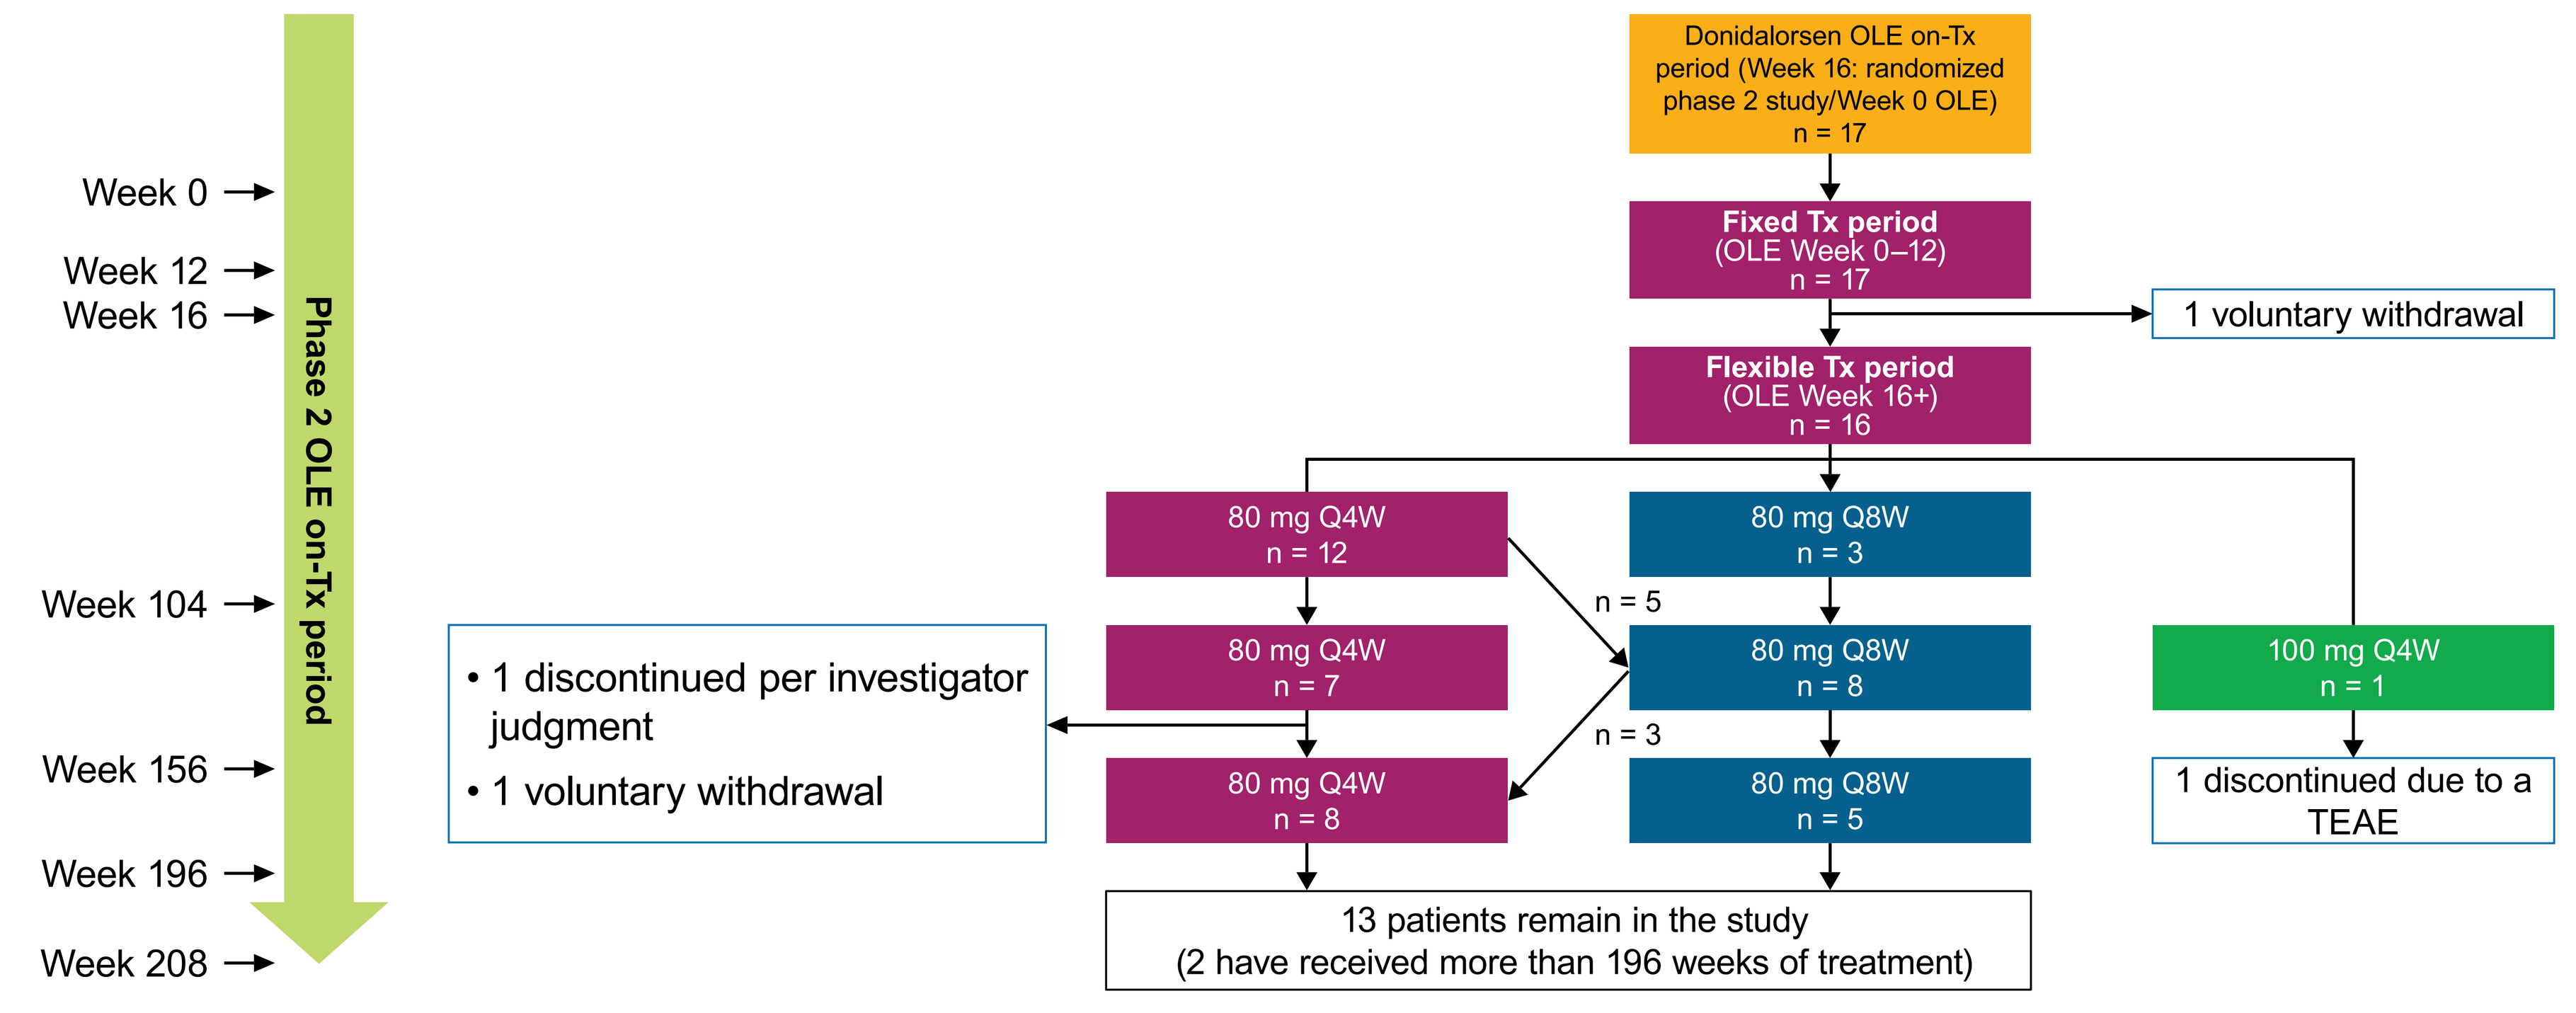
Week 0
Week 12
Week 16
Week 104
Week 156
Week 196
Week 208
Phase 2 OLE on-Tx period
Donidalorsen OLE on-Tx period (Week 16: randomized phase 2 study/Week 0 OLE) n = 17
Fixed Tx period (OLE Week 0–12) n = 17
1 voluntary withdrawal
Flexible Tx period (OLE Week 16+) n = 16
80 mg Q4W n = 12
80 mg Q8W n = 3
n = 5
80 mg Q4W n = 7
80 mg Q8W n = 8
100 mg Q4W n = 1
n = 3
80 mg Q4W n = 8
80 mg Q8W n = 5
1 discontinued due to a TEAE
• 1 discontinued per investigator judgment
• 1 voluntary withdrawal
13 patients remain in the study
(2 have received more than 196 weeks of treatment)

OLE, open-label extension; Q4W, once every 4 weeks; Q8W, once every 8 weeks; TEAE, treatment-emergent adverse event; Tx, treatment.

- A total of 17 patients enrolled in the OLE study and 4 have discontinued
- Of these, 8 patients have received donidalorsen Q8W

Table 1. Overview of TEAEs

	Donidalorsen (N = 17)
Any TEAE,[a] n (%)	15 (88)
Related to study drug	7 (41)
Mild	3 (18)
Moderate	4 (24)
Severe	0
Leading to discontinuation	1[b] (6)
Any serious TEAE,[a] n (%)	0
Most common TEAEs[a] (≥15% of patients), n (%)	
COVID-19	8 (47)
Urinary tract infection	4 (24)
Headache	4 (24)
Gastroenteritis viral	3 (18)
Influenza	3 (18)
Nasopharyngitis	3 (18)
Abdominal pain	3 (18)
Nausea	3 (18)
Injection-site discoloration	3 (18)
Cough	3 (18)

[a]TEAE is defined as any adverse event starting or worsening on or after the first dose of donidalorsen. [b]Injection-site discoloration events led to discontinuation in 1 patient in the 100 mg Q4W group.
COVID-19, coronavirus disease 2019; Q4W, once every 4 weeks; TEAE, treatment-emergent adverse event.

- Of the 15 patients with TEAEs, 12 (80%) reported mild or moderate TEAEs
- There were no serious TEAEs and no severe TEAEs related to the study drug
- The most common TEAEs were coronavirus disease 2019 (n = 8), urinary tract infection (n = 4), and headache (n = 4)
- The most common TEAE related to the study drug was injection-site discoloration (n = 3)

Figure 3. Summary of HAE Attack Rates

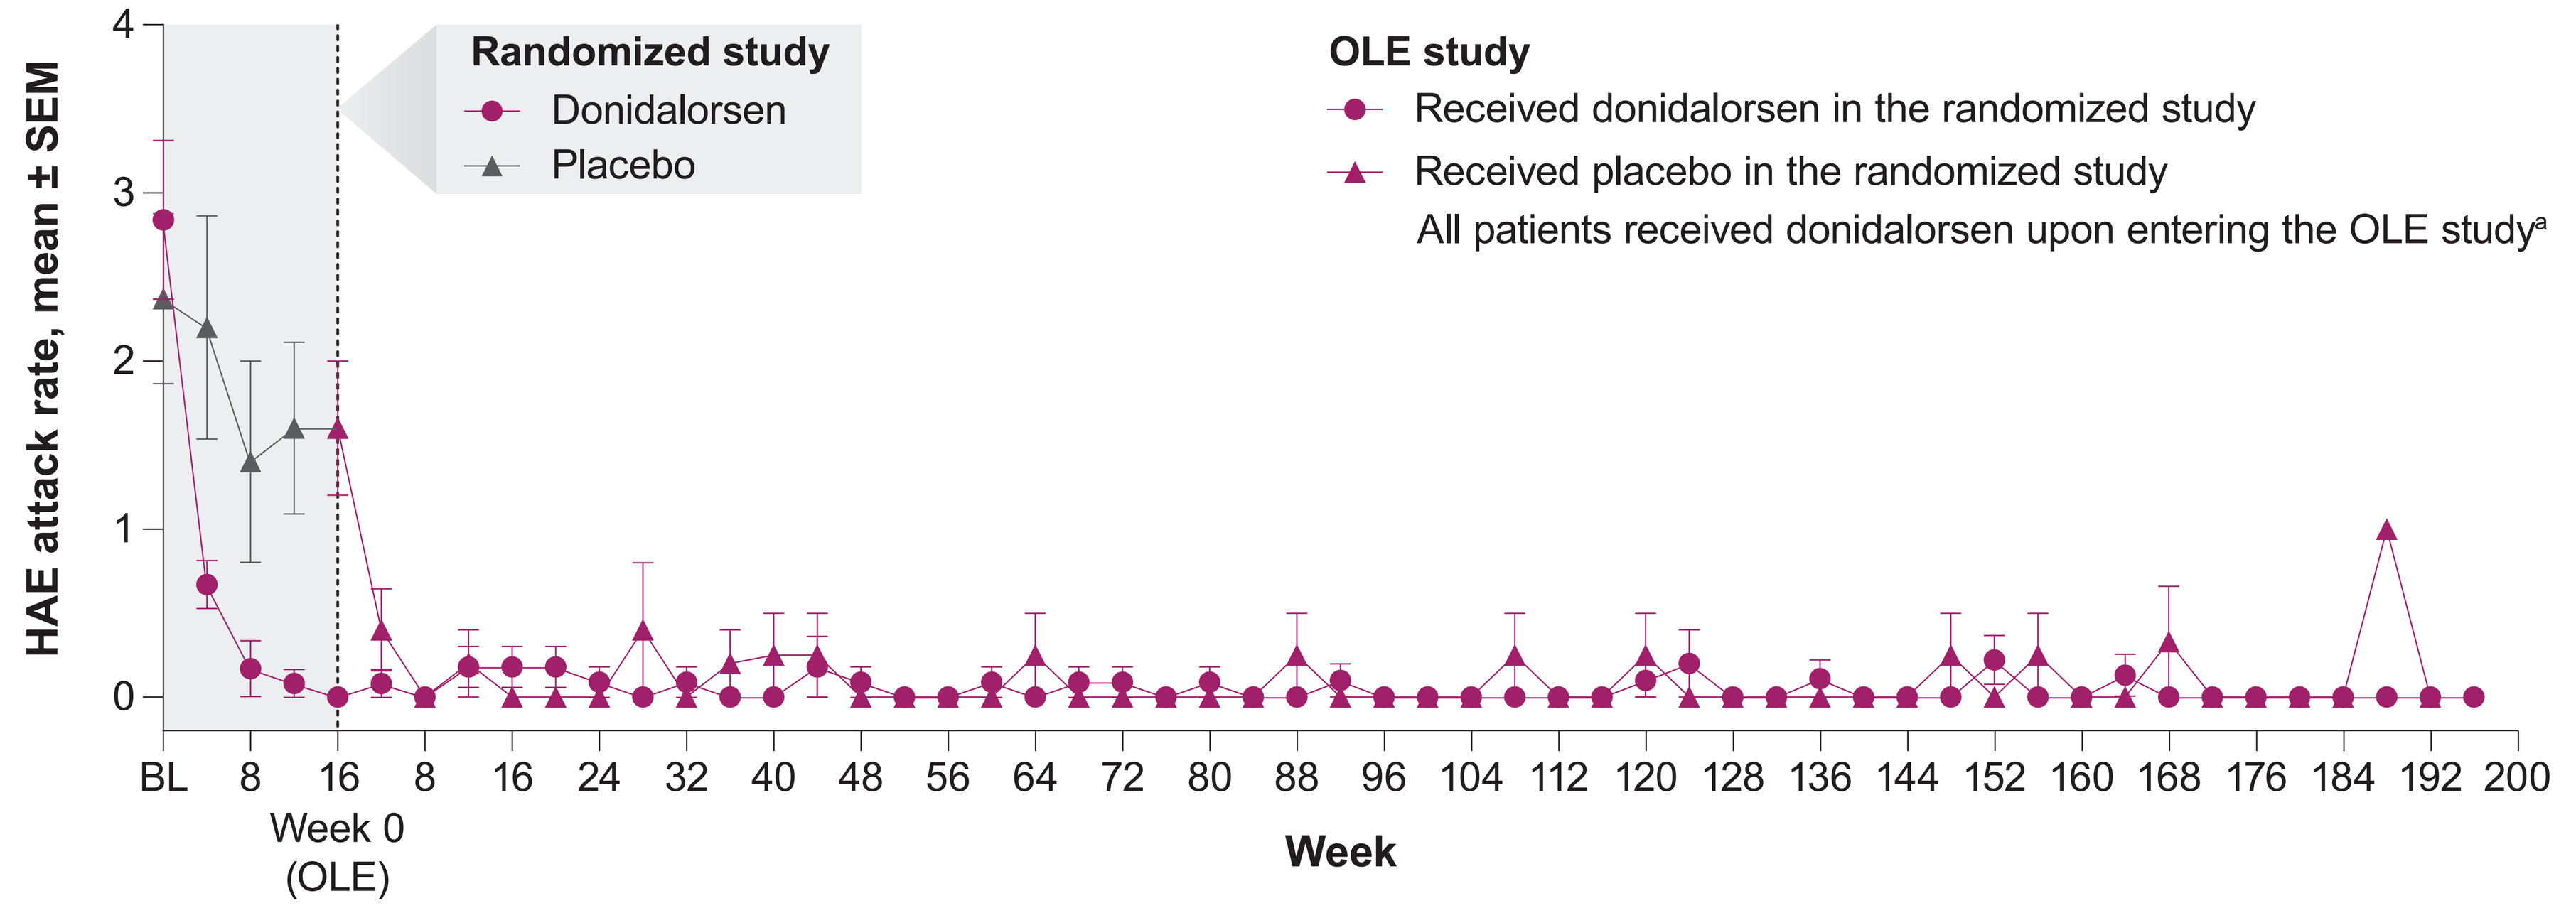
HAE attack rate, mean ± SEM
Randomized study
Donidalorsen
Placebo
OLE study
Received donidalorsen in the randomized study
Received placebo in the randomized study
All patients received donidalorsen upon entering the OLE study[a]
BL
Week 0 (OLE)
Week

BL was defined as the 56-day run-in period of the phase 2 randomized study.
[a]Data are pooled across patients in all dosing groups during the flexible dosing period (donidalorsen: 80 mg Q4W, 80 mg Q8W, or 100 mg Q4W).
BL, baseline; HAE, hereditary angioedema; OLE, open-label extension; Q4W, once every 4 weeks; Q8W, once every 8 weeks; SEM, standard error of the mean.

- Mean monthly HAE attack rate decreased by 96% from baseline among all patients during the on-treatment period over 196 weeks
 - Mean (standard error of the mean) monthly attack rates decreased from 2.70 (0.36) to 0.06 (0.02)
 - Mean decreases from baseline to the flexible dosing period were 97% in the 80 mg Q4W group and 83% in the Q8W group

Figure 4. AE-QoL Scores at Baseline and Week 156

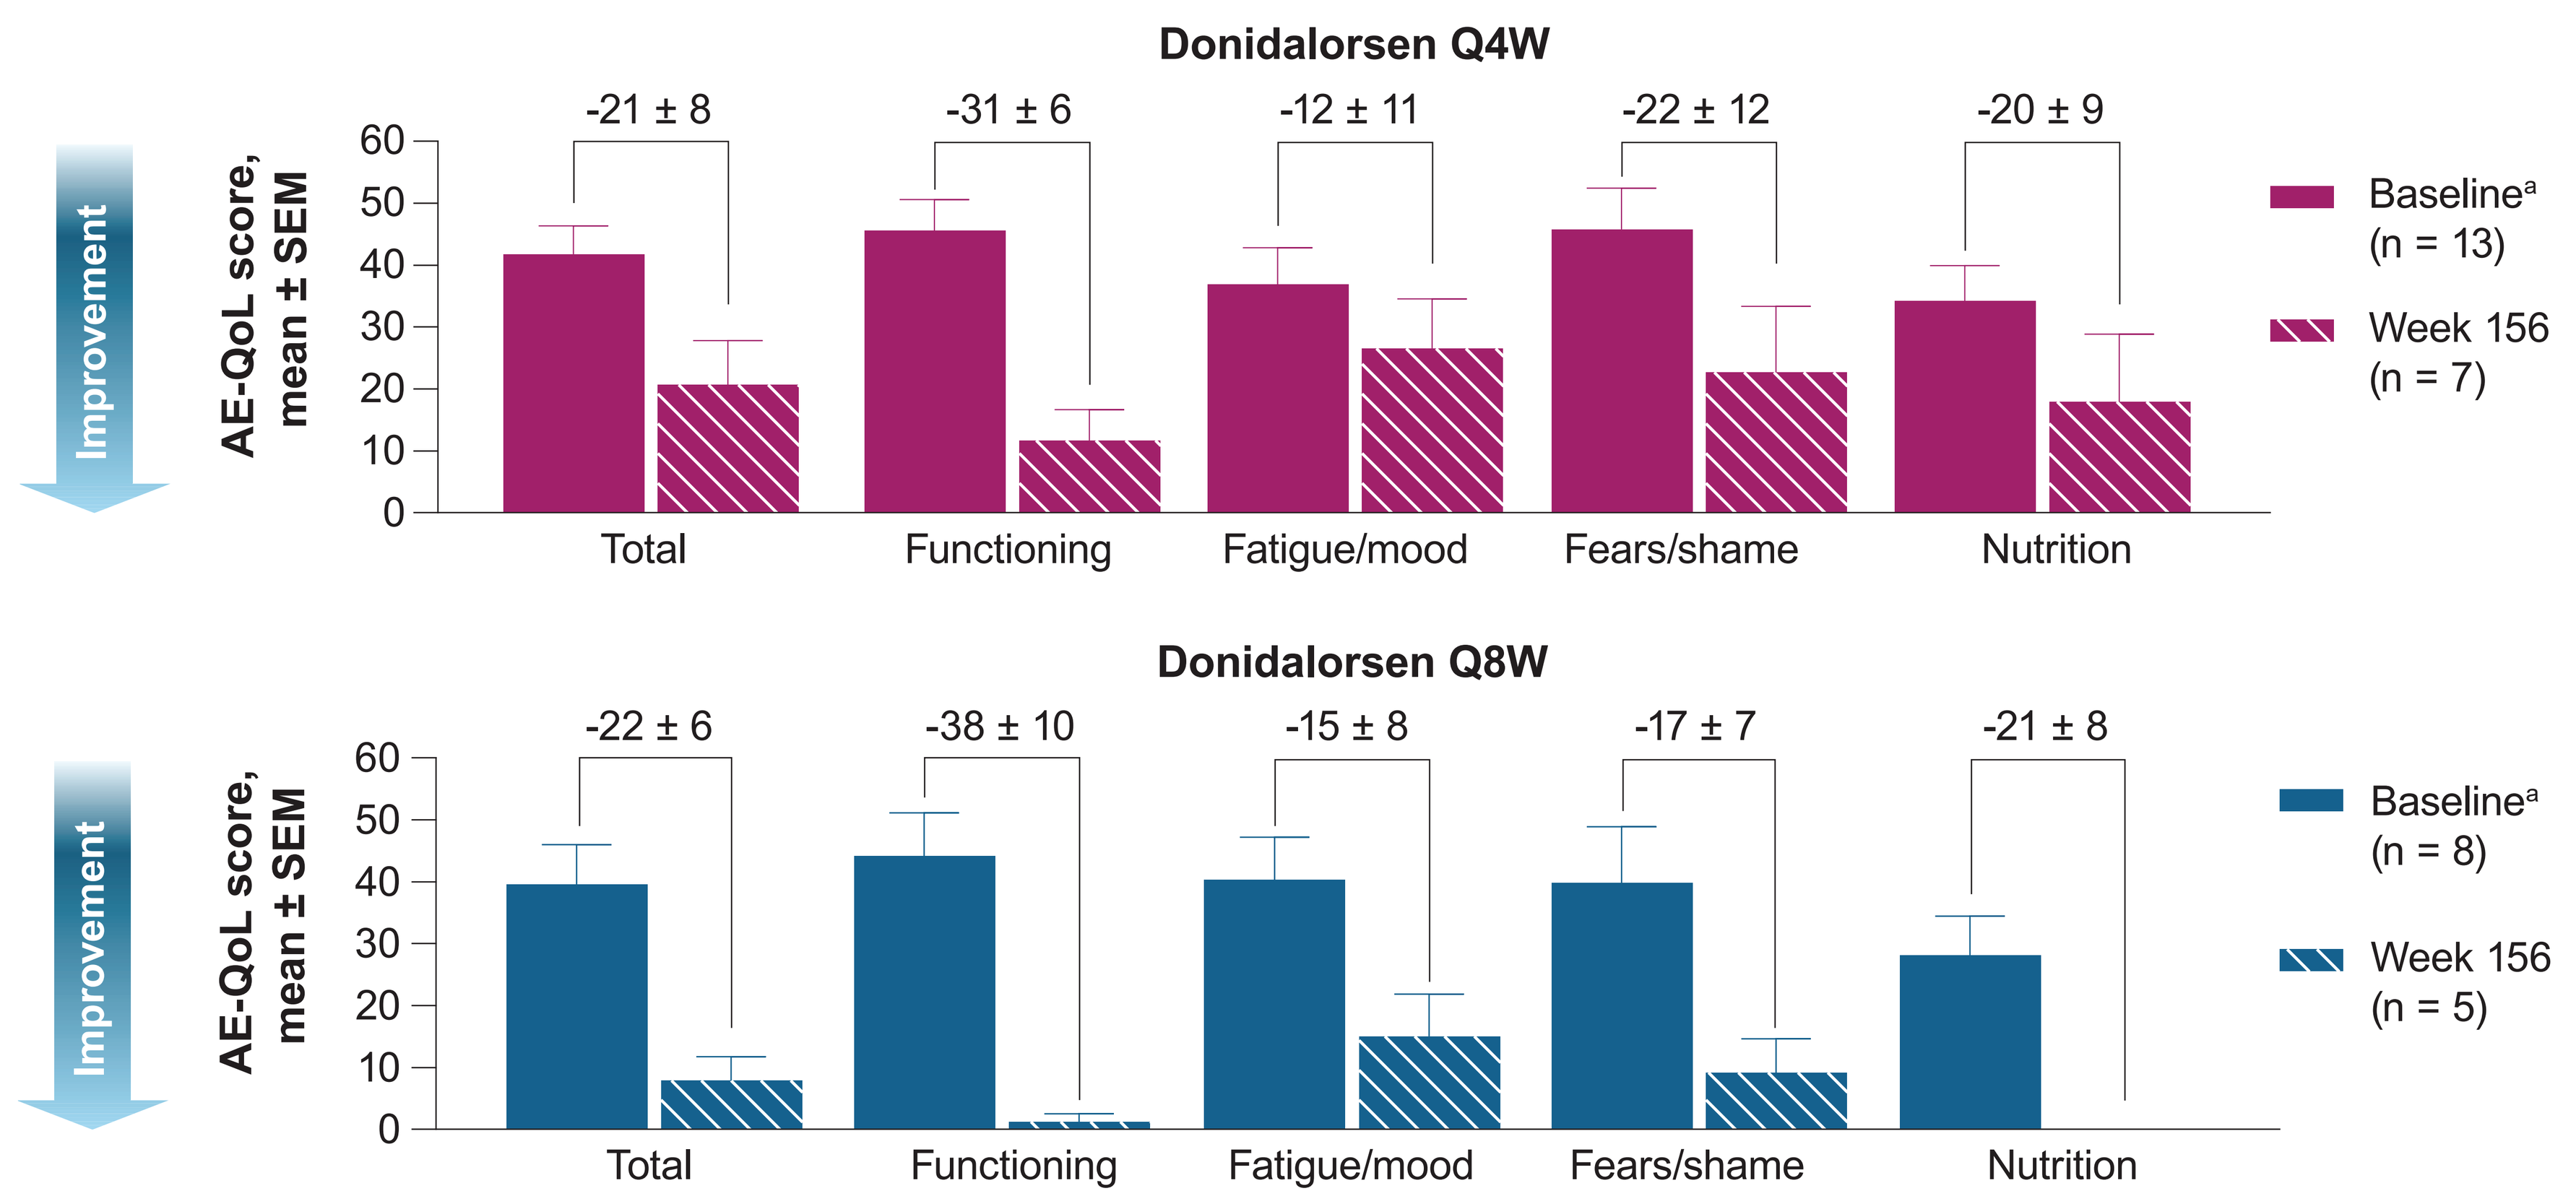
Donidalorsen Q4W
Improvement
AE-QoL score, mean ± SEM
-21 ± 8
-31 ± 6
-12 ± 11
-22 ± 12
-20 ± 9
Total
Functioning
Fatigue/mood
Fears/shame
Nutrition
Baseline[a] (n = 13)
Week 156 (n = 7)
Donidalorsen Q8W
-22 ± 6
-38 ± 10
-15 ± 8
-17 ± 7
-21 ± 8
Baseline[a] (n = 8)
Week 156 (n = 5)

[a]Baseline is defined as baseline of the phase 2 randomized study.
AE-QoL, Angioedema Quality of Life Questionnaire; Q4W, once every 4 weeks; Q8W, once every 8 weeks; SEM, standard error of the mean.

- The mean AE-QoL total score decreased by 21 points overall in patients receiving donidalorsen Q4W from baseline to the prespecified Week 156 time point
 - The mean decrease was 22 points in patients who received donidalorsen Q8W
- Patients receiving donidalorsen Q4W or Q8W reported improvements in all AE-QoL domains

Conclusions

Safety and Tolerability

- Over 196 weeks, donidalorsen had an acceptable safety and tolerability profile

Efficacy

- Donidalorsen led to a sustained reduction in HAE attack rate for up to 196 weeks

Quality of Life

- Patients receiving donidalorsen Q4W or Q8W reported improvements in all AE-QoL domains from baseline to Week 156

ACKNOWLEDGMENTS

We thank the study participants and their families and the team of investigators, research coordinators, and study staff. We also thank Tracy Reigle, of Ionis, for graphic art assistance with the figures and Nayna Sanathara, PhD, of Ionis, for editorial review. Medical writing and editorial assistance were provided by Jake Wilmot, PhD, of Red Nucleus and funded by Ionis Pharmaceuticals, Inc. This study was funded by Ionis Pharmaceuticals, Inc.

DISCLOSURES

MM has received research support from Astria, BioCryst, BioMarin, CSL Behring, Ionis, KalVista, Pharming, Pharvaris, and Takeda; consultancy fees from BioCryst, CSL Behring, KalVista, Pharming, and Takeda; and speaking fees from BioCryst, CSL Behring, Pharming, and Takeda. **LB**, **YD**, **ST**, and **KBN** are employees of Ionis and hold shares and/or options in Ionis. **DMC** has received speaker or consultancy fees from Astria, BioCryst, CSL Behring, Intellia, Ionis, KalVista, Pharvaris, and Takeda. **MAR** has received research grants from BioCryst, BioMarin Pharmaceutical, CSL Behring, Ionis, KalVista, Pharvaris, and Takeda; consulted for Astria, BioCryst, BioMarin Pharmaceutical, Celldex, CSL Behring, Cycle Pharma, Grifols, Intellia Therapeutics, Ionis, KalVista, Pfizer, Pharming, Pharvaris, Sanofi-Regeneron, and Takeda; and provided speaker presentations for CSL Behring, Grifols, Pharming, and Takeda. **RT** has received grants or research support from Astria, BioCryst, CSL Behring, Ionis, KalVista, Pharvaris, and Takeda; is a speaker for AstraZeneca, BioCryst, CSL Behring, Grifols, GSK, Pharming, Sanofi-Regeneron, and Takeda; and has served as a consultant for BioCryst, CSL Behring, KalVista, Pharming, and Takeda.

REFERENCES

1. Riedl MA, et al. *J Allergy Clin Immunol Pract*. 2024;12(4):911-8.
2. Raasch J, et al. *World Allergy Organ J*. 2023;16(6):100792.
3. Busse P, et al. *J Allergy Clin Immunol Pract*. 2022;10(3):716-22.
4. Riedl MA, et al. *N Engl J Med*. 2024;391(1):21-31.
5. Fijen LM, et al. *N Engl J Med*. 2022;386(11):1026-33.

American Academy of Allergy, Asthma, and Immunology/World Allergy Organization Joint Congress • San Diego, CA, USA • February 28–March 3, 2025 • Poster 654

michaelmanningmd@aol.com